CAR-TS ARMORED WITH SMALL MOLECULE-REGULATED IL12 OR CD40L CASSETTES FOR ENHANCED ACTIVITY AGAINST SOLID TUMORS

OBSIDIAN™
1030 Massachusetts Ave, Cambridge, MA 02138

LB-013 Clinical Research 1/ Endocrinology

Michelle Ols, Michael Schebesta, Emily Brideau, Kutlu Elpek, Michelle Fleury, Jennifer Gori, Scott Heller, Dan Jun Li, Benjamin Primack, Christopher Reardon, Dhruv Sethi, Alex Storer, Dexue Sun, Karen Tran, Elizabeth Weisman, Michael Briskin, Celeste Richardson, Vipin Suri, and **Steven Shamah**

ABSTRACT

Adoptive cell therapy with chimeric antigen receptor (CAR) modified T cells has demonstrated remarkable clinical efficacy in the treatment of certain B cell malignancies, and more recently in multiple myeloma. However, CAR-T therapy has been less successful in treating solid tumors due to multiple obstacles, including the lack of robust CAR-T cell expansion, the immunosuppressive tumor microenvironment, and tumor escape due to the loss of targeted antigen. Engineering CAR-T cells to produce immunomodulatory factors such as Interleukin 12 (IL12) and Cluster of Differentiation 40 Ligand (CD40L) has been shown to enhance functional activity by driving T cell expansion, conferring resistance to immunosuppression, improving antigen presentation, and inducing antigen spread. However, the clinical utility of both IL12 and activators of the CD40 signaling pathway have been limited by systemic toxicity associated with their potent pharmacological activities. We describe here the implementation of ligand-controlled regulation of IL12 and CD40L *in vitro* and *in vivo* in engineered primary human T cells via the use of destabilizing domain (DD) technology. DDs are small protein domains that are misfolded and inherently unstable in the cell, but which can be reversibly stabilized by the binding of approved pharmacologic agents. This conditional stability of DDs can be readily conferred to any protein of interest by fusing it to the DD, thus providing fine-tuned, exogenous regulation of protein expression and function. We show that transduction of human T cells with either DD-IL12 or DD-CD40L fusion constructs yields low expression levels in the basal state and a rapid, dose-dependent induction of IL12 or CD40L protein in the presence of stabilizing ligand. Moreover, kinetically precise, on-demand production of either factor from CAR-T cells can be achieved in mice by oral dosing. Providing precise tuning of the timing and level of expression of these immunomodulatory factors in CAR-T cells could significantly enhance safety and therapeutic efficacy, in particular against solid tumor malignancies.

Figure 1: The Dramatic Success of CAR-Ts has Reinvigorated the Field of Cell Therapy but Substantial Challenges Remain

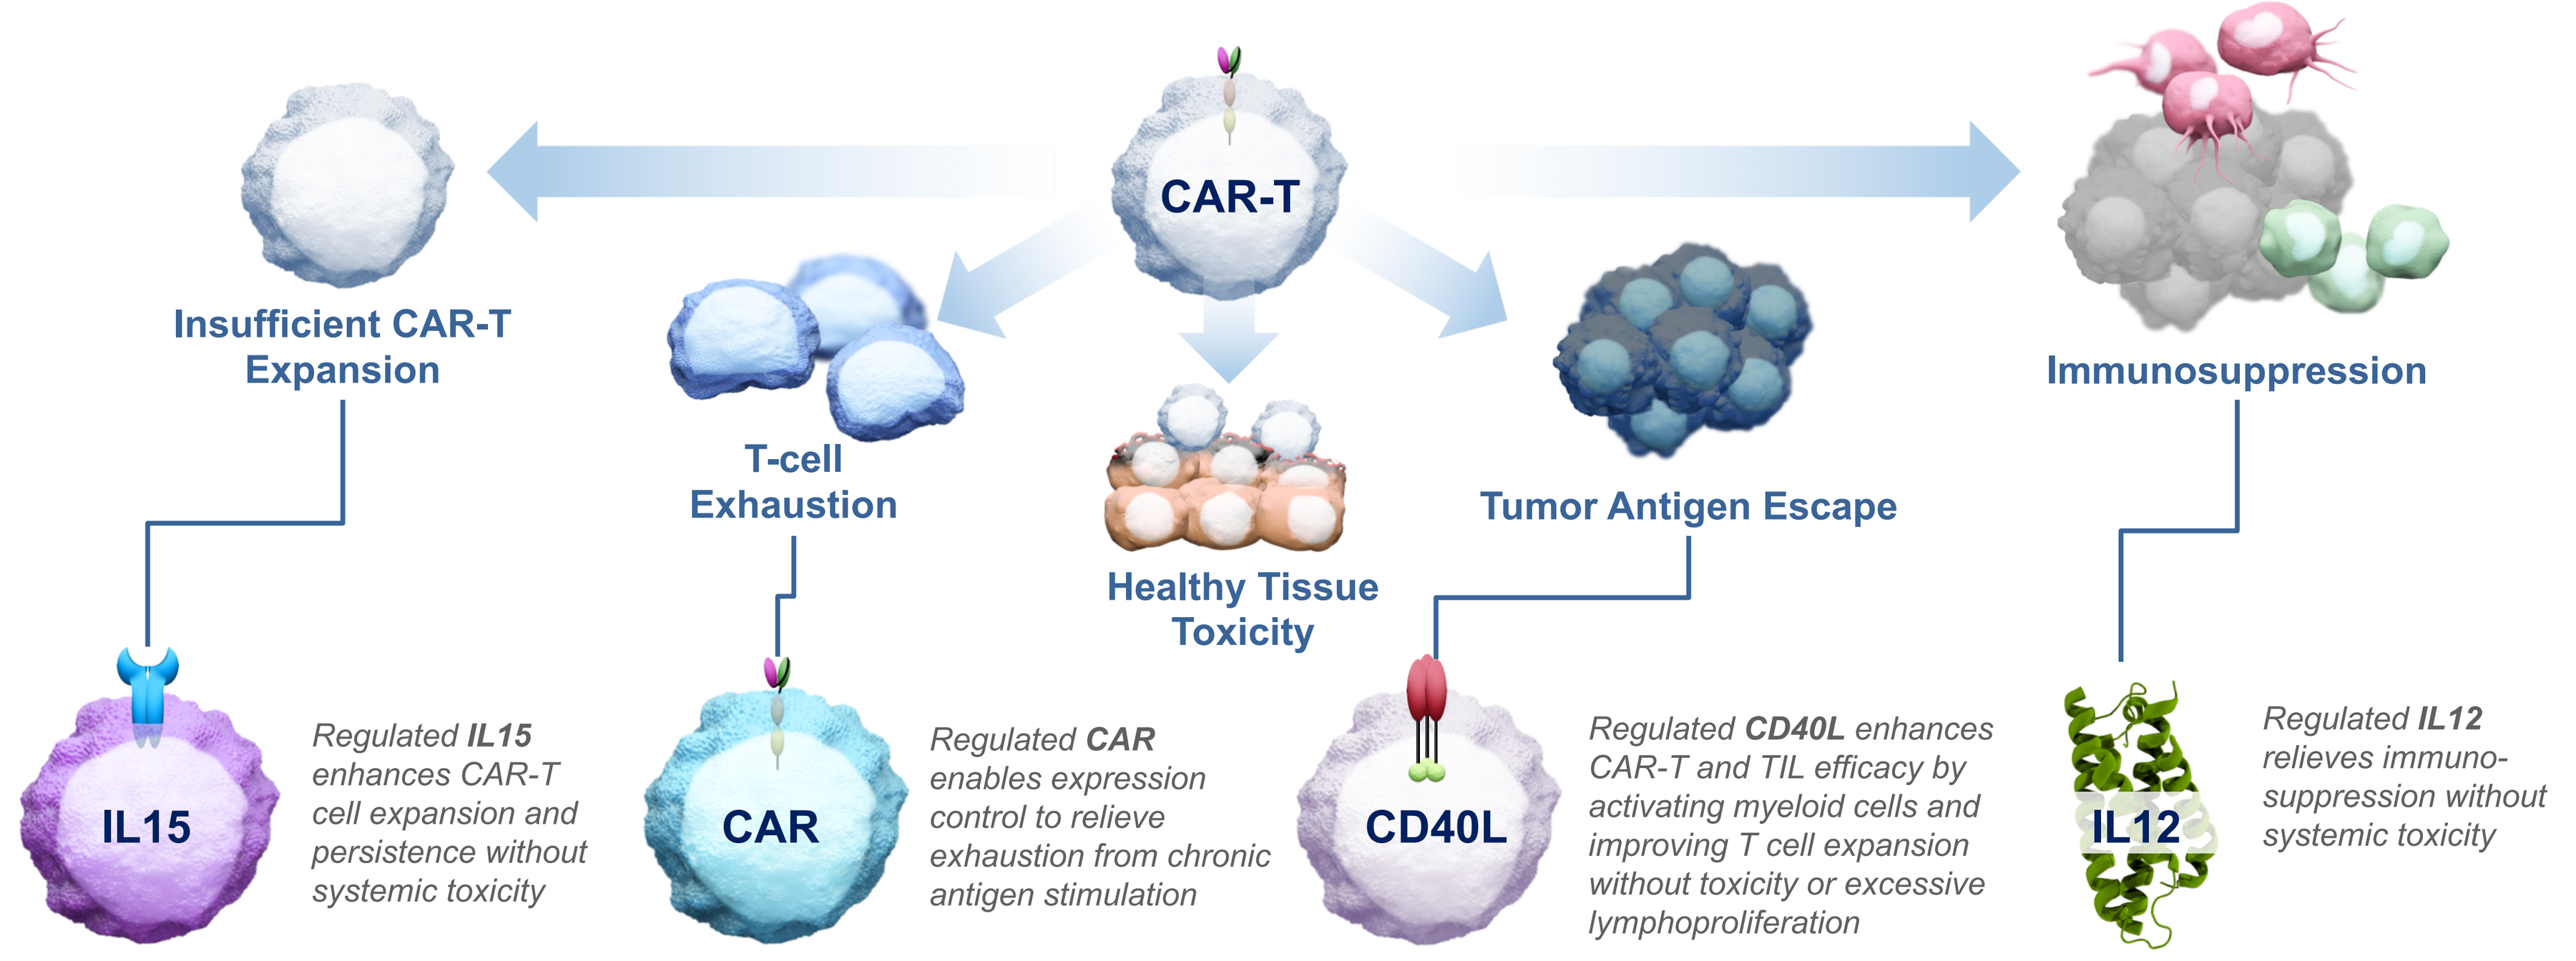
CAR-T
Insufficient CAR-T Expansion
T-cell Exhaustion
Healthy Tissue Toxicity
Tumor Antigen Escape
Immunosuppression
IL15
Regulated *IL15* enhances CAR-T cell expansion and persistence without systemic toxicity
CAR
Regulated *CAR* enables expression control to relieve exhaustion from chronic antigen stimulation
CD40L
Regulated *CD40L* enhances CAR-T and TIL efficacy by activating myeloid cells and improving T cell expansion without toxicity or excessive lymphoproliferation
IL12
Regulated *IL12* relieves immuno-suppression without systemic toxicity

Many of these challenges could be addressed by potent cytokine/co-stimulatory molecules whose constitutive activation would also provide significant toxicity concerns. Obsidian is developing cell therapy products armored with immunomodulatory factors whose expression can be carefully titrated for safe and more effective therapeutics.

Figure 2: Small Molecule Regulated Protein Expression Using Destabilizing Domains

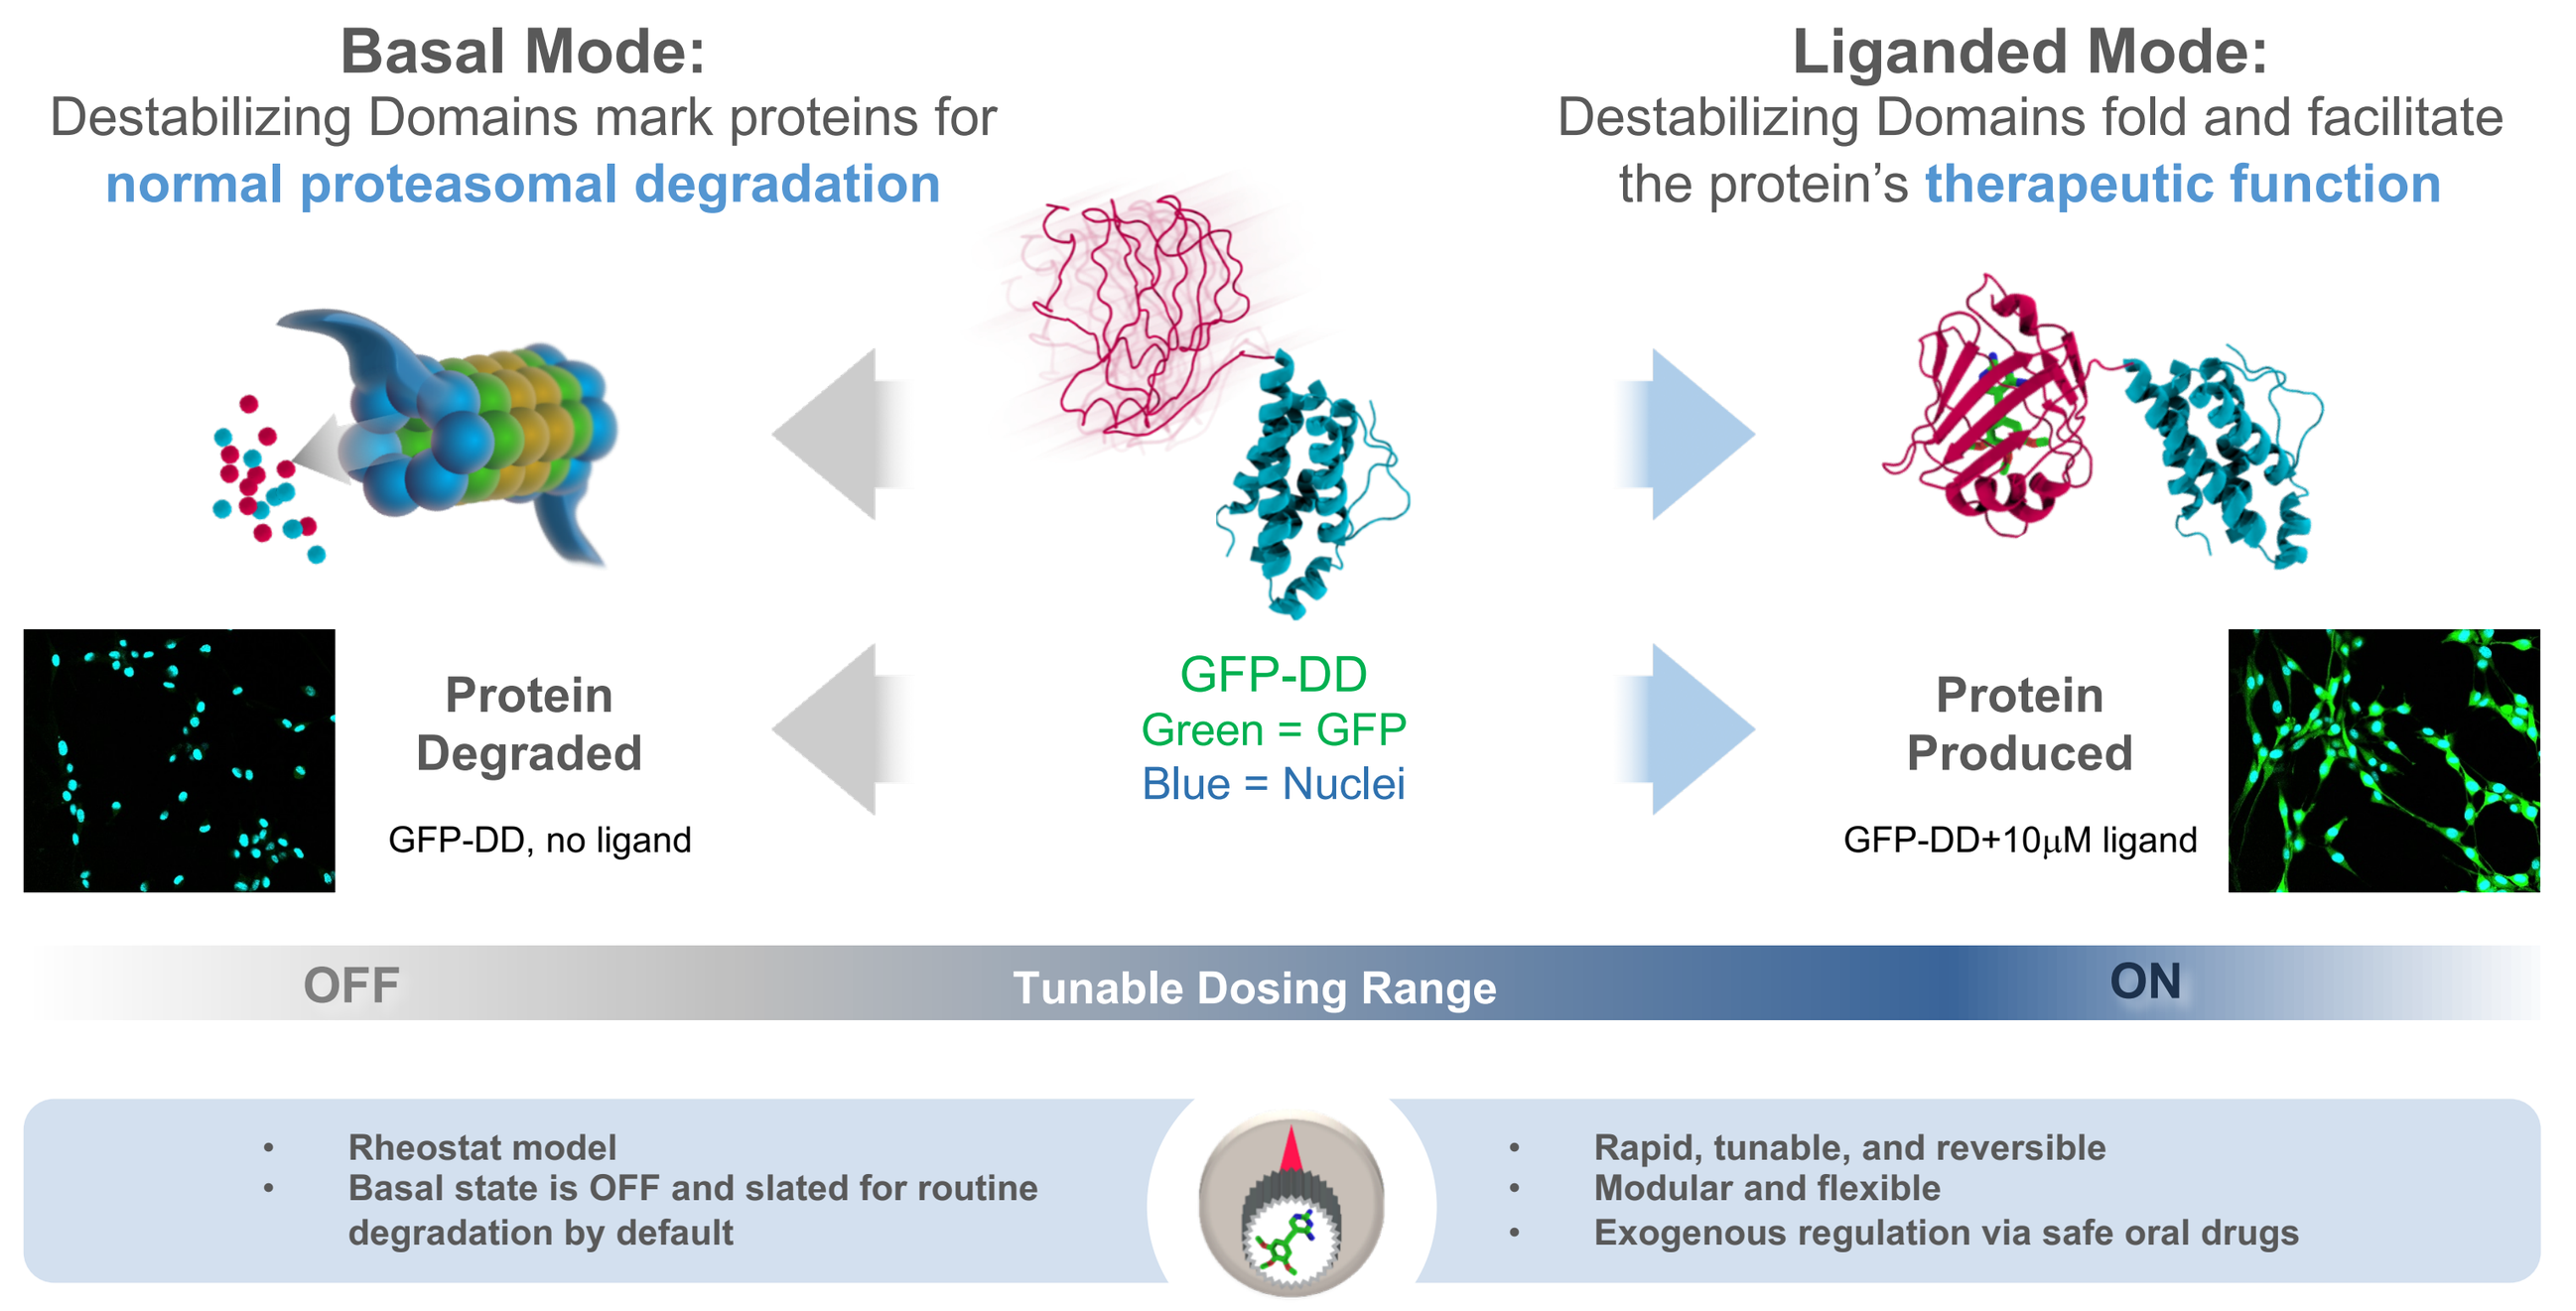
Basal Mode:
Destabilizing Domains mark proteins for normal proteasomal degradation
Liganded Mode:
Destabilizing Domains fold and facilitate the protein's therapeutic function
Protein Degraded
GFP-DD, no ligand
GFP-DD
Green = GFP
Blue = Nuclei
Protein Produced
GFP-DD+10μM ligand
OFF
Tunable Dosing Range
ON
Rheostat model
Basal state is OFF and slated for routine degradation by default
Rapid, tunable, and reversible
Modular and flexible
Exogenous regulation via safe oral drugs

DDs Used in this Study

Destabilizing Domain (DD)	Ligand
E.Coli dihydrofolate reductase (ecDHFR)	Trimethoprim (TMP)
Estrogen receptor (huER)	Bazedoxifene
Carbonic anhydrase 2 (huCA2)	Acetazolamide

The Obsidian technology provides control over protein stability via the administration of safe, FDA-approved small molecule drugs. Destabilizing Domains (DDs) are expressed as unfolded and unstable units that confer rapid degradation to fused proteins through the cell's proteasome machinery. The binding of a small molecule ligand to the DD stabilizes the complex enabling the expression and function of the target gene product. Importantly, the stabilization is titratable based on the concentration of dosed ligand providing fine-tuned control over protein function.

Figure 3: CD40L Expression Enhances Anti-Tumor Efficacy of CD19 CAR-T Cells in Acute Lymphoblastic Leukemia Model

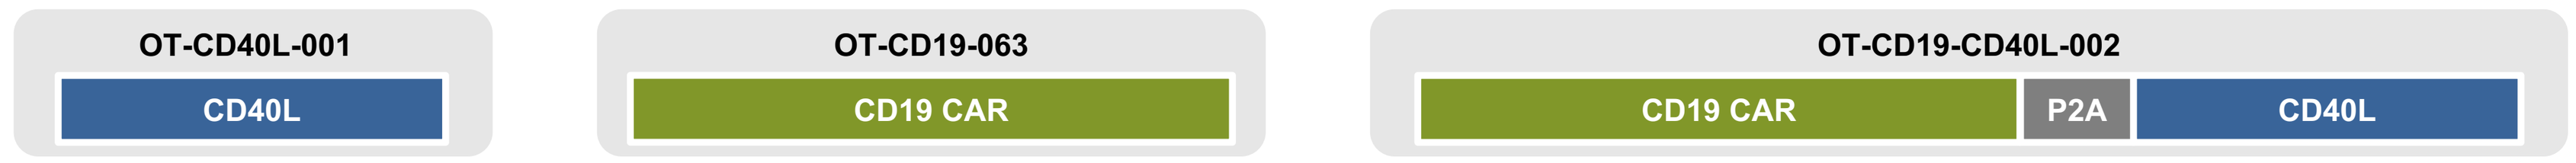
OT-CD40L-001
CD40L
OT-CD19-063
CD19 CAR
OT-CD19-CD40L-002
CD19 CAR
P2A
CD40L

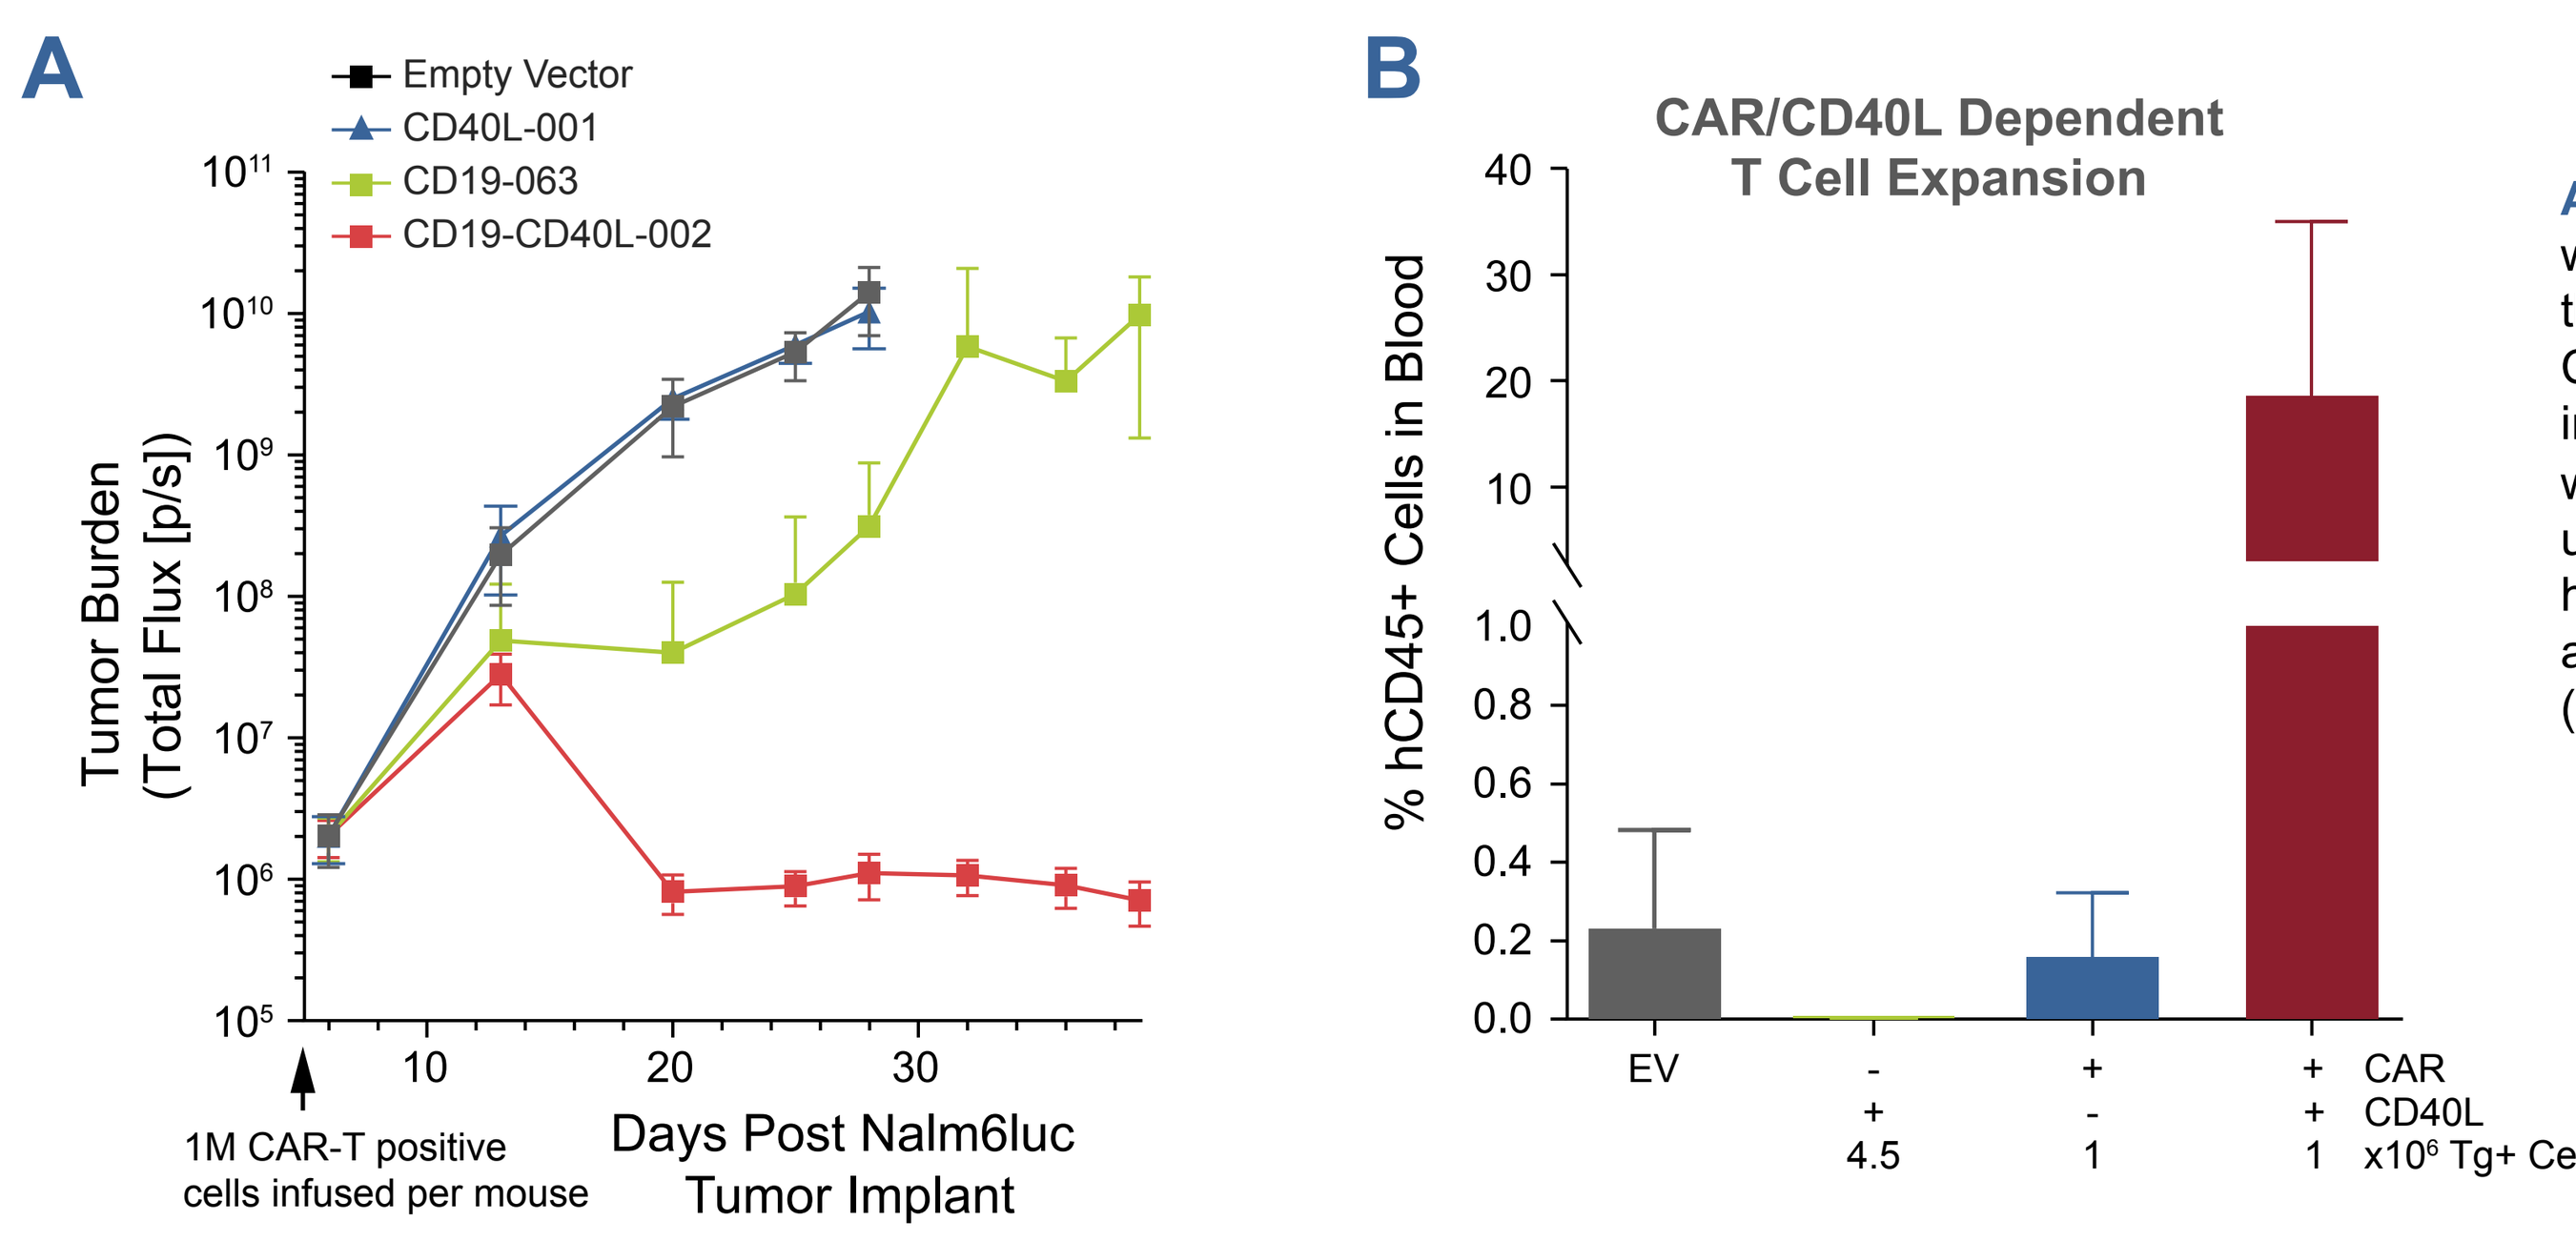
A
Empty Vector
CD40L-001
CD19-063
CD19-CD40L-002
Tumor Burden (Total Flux [p/s])
1M CAR-T positive cells infused per mouse
Days Post Nalm6luc Tumor Implant
B
CAR/CD40L Dependent T Cell Expansion
% hCD45+ Cells in Blood
EV
CAR
CD40L
x10⁶ Tg+ Cells

A) Nalm6-luc tumor-bearing NSG mice were injected with T cells transduced with the indicated constructs at 10^6 CAR⁺ or CD40L⁺ cells per mouse 7 days post tumor implantation (n=8/group). Tumor burden was quantified using bioluminescence upon luciferin administration. **B**) Circulating human T cells were quantified using CD45 as a FACS marker at day 21 of the study (14 days after T cell infusion).

Figure 4: Regulation of CD40L Levels and Function *in vitro* and *in vivo*

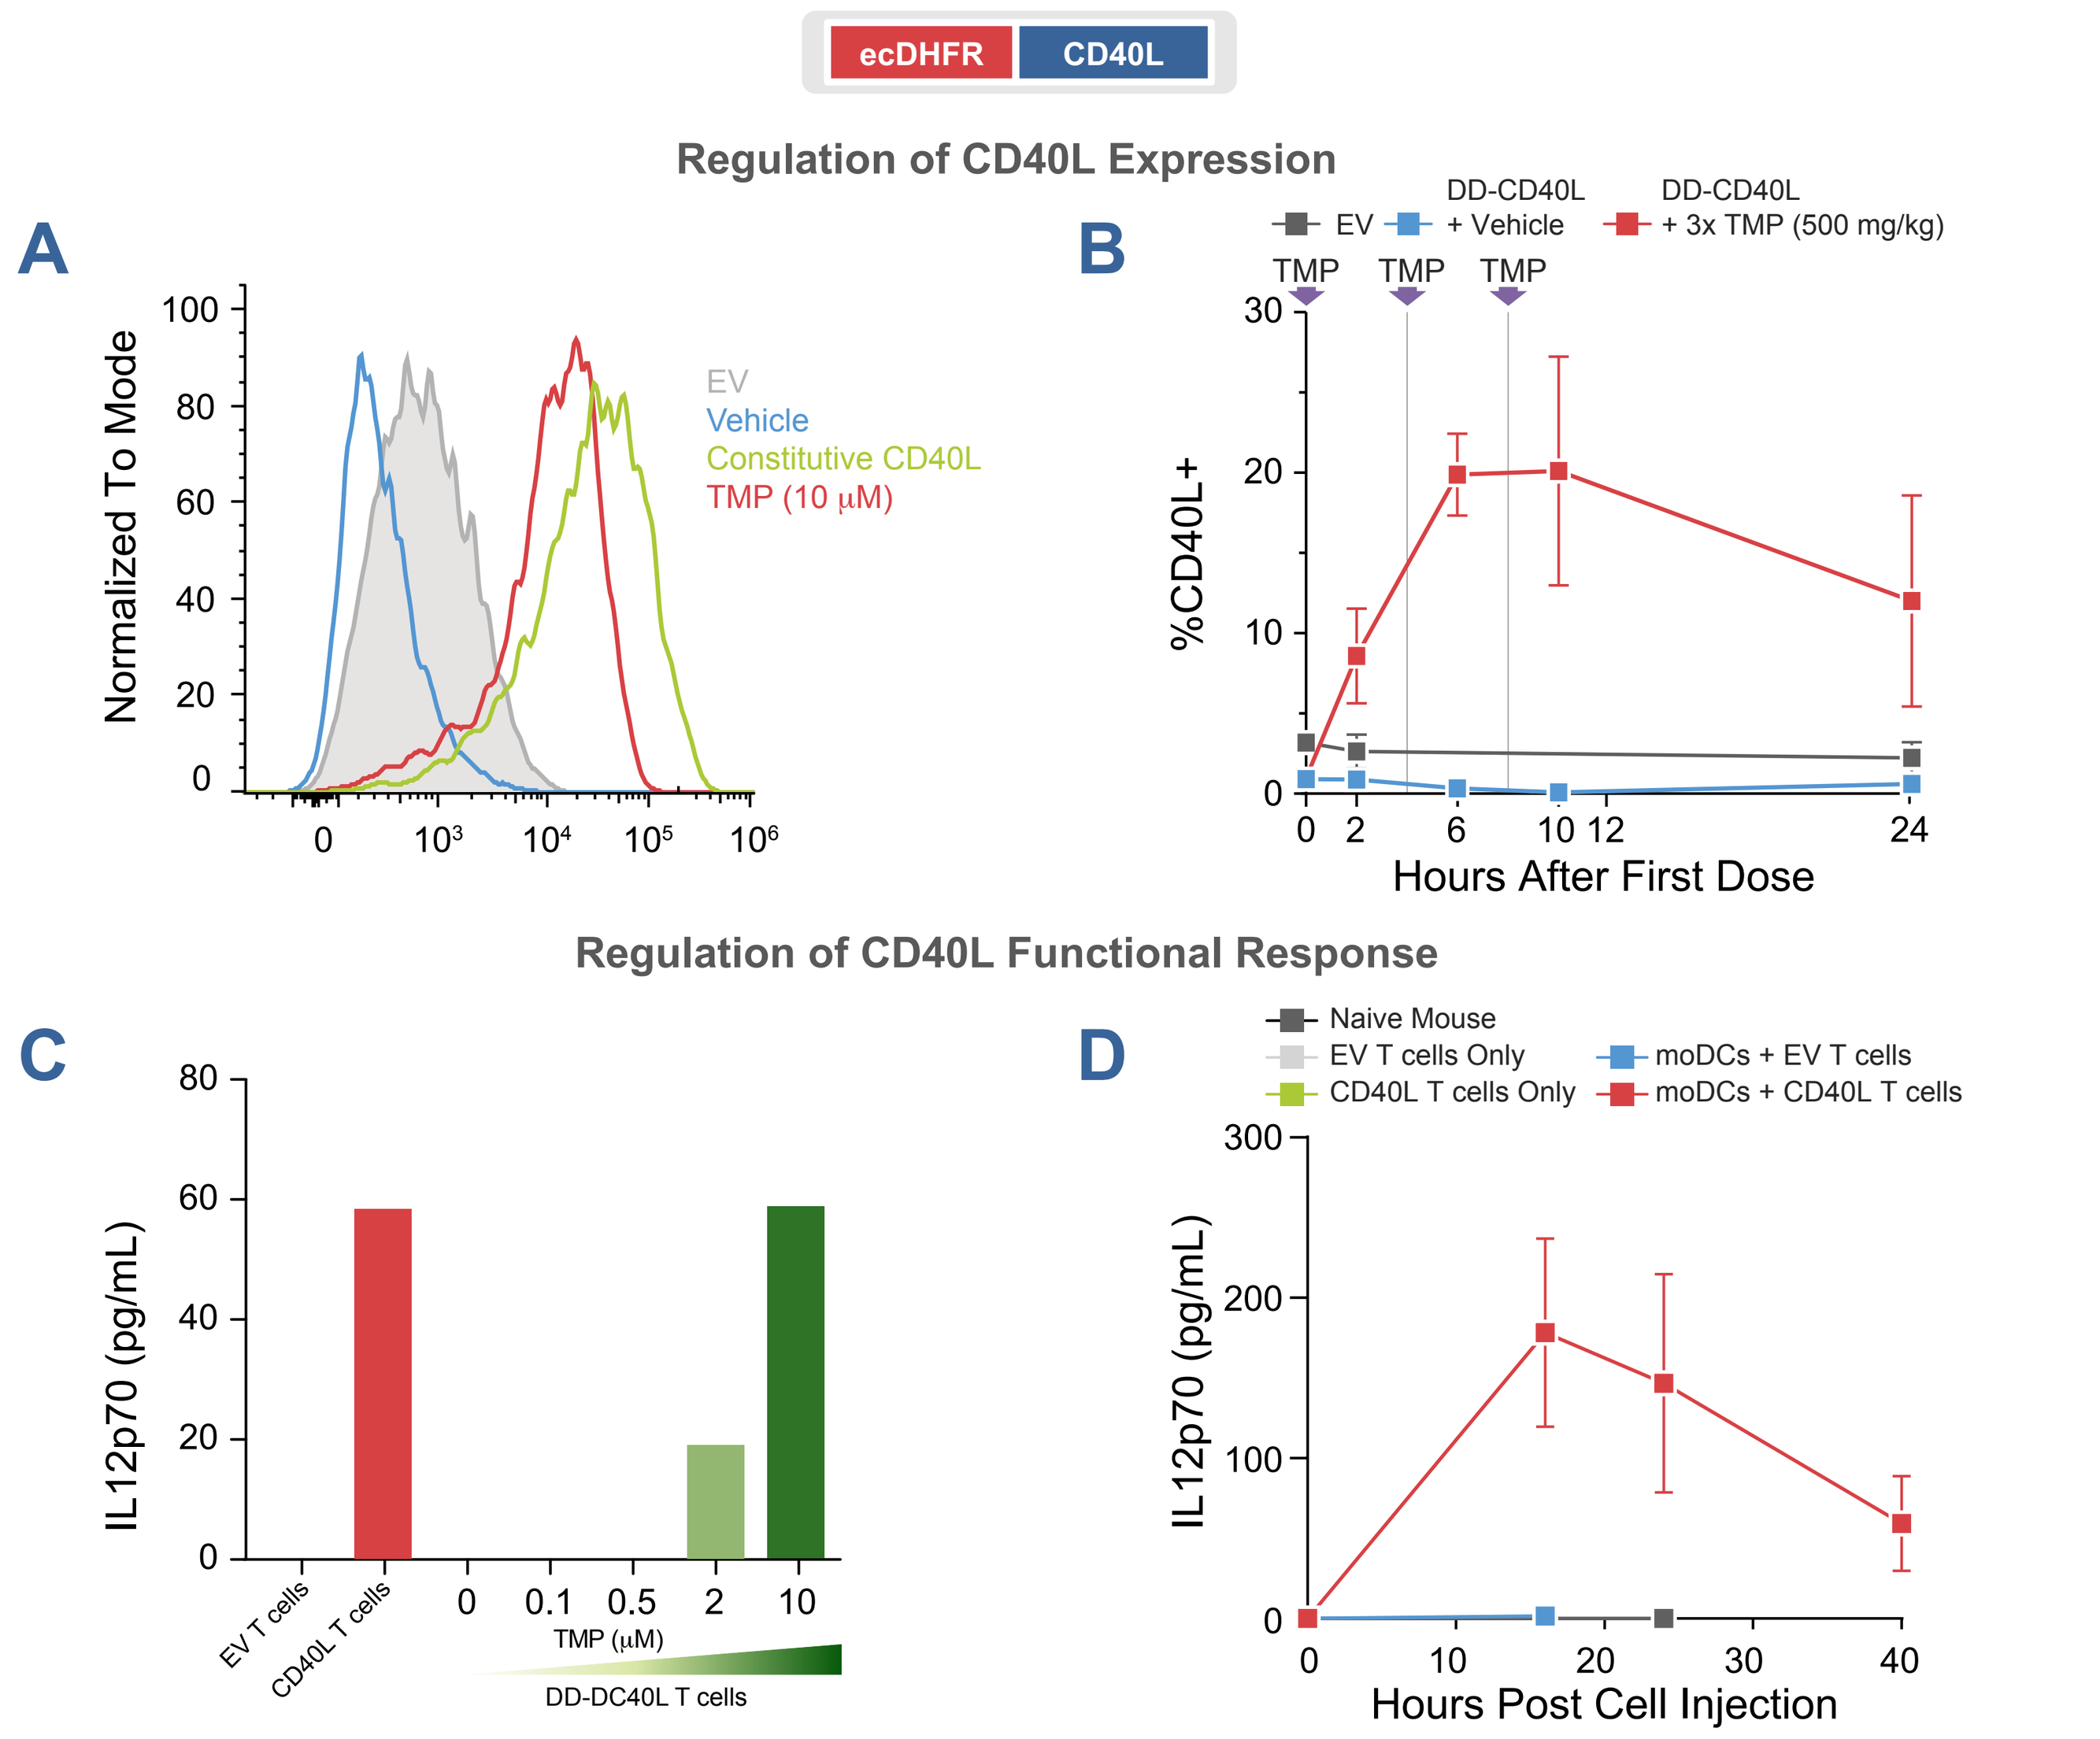
ecDHFR
CD40L
Regulation of CD40L Expression
A
Normalized To Mode
EV
Vehicle
Constitutive CD40L
TMP (10 μM)
B
EV
DD-CD40L + Vehicle
DD-CD40L + 3x TMP (500 mg/kg)
TMP
%CD40L+
Hours After First Dose
Regulation of CD40L Functional Response
C
IL12p70 (pg/mL)
EV T cells
CD40L T cells
TMP (μM)
DD-DC40L T cells
D
Naive Mouse
EV T cells Only
CD40L T cells Only
moDCs + EV T cells
moDCs + CD40L T cells
IL12p70 (pg/mL)
Hours Post Cell Injection

A) Activated T cells were transduced with constitutive or E. coli DHFR regulated CD40L. Two days later, cells were treated with vehicle (DMSO) or TMP for 24h and analyzed for CD40L surface expression. **B**) Transduced T cells were expanded *in vitro* for 10 days before infusion into NSG mice (n=4/group). Two days after infusion animals were dosed orally at the indicated times with 500 mg/kg TMP (▼). Blood samples taken prior to dosing and at the indicated post-dosing time points were analyzed for CD40L surface expression by FACS. **C**) Transduced T cells were co-cultured (10:1 ratio) with allogeneic human monocytes differentiated into dendritic cells with IL4 and GM-CSF for 5 days before cryopreservation. Secreted IL12 was measured by MSD assay after two days of co-culture in the absence or presence of TMP. **D**) Transduced T cells and human dendritic cells were freshly thawed and separately injected intraperitonially at a 5:1 ratio into NSG mice. Plasma was collected at the indicated timepoints post cell infusion and analyzed for the presence of IL12.

Figure 5: Regulation of CD40L with a Clinically Translatable DD

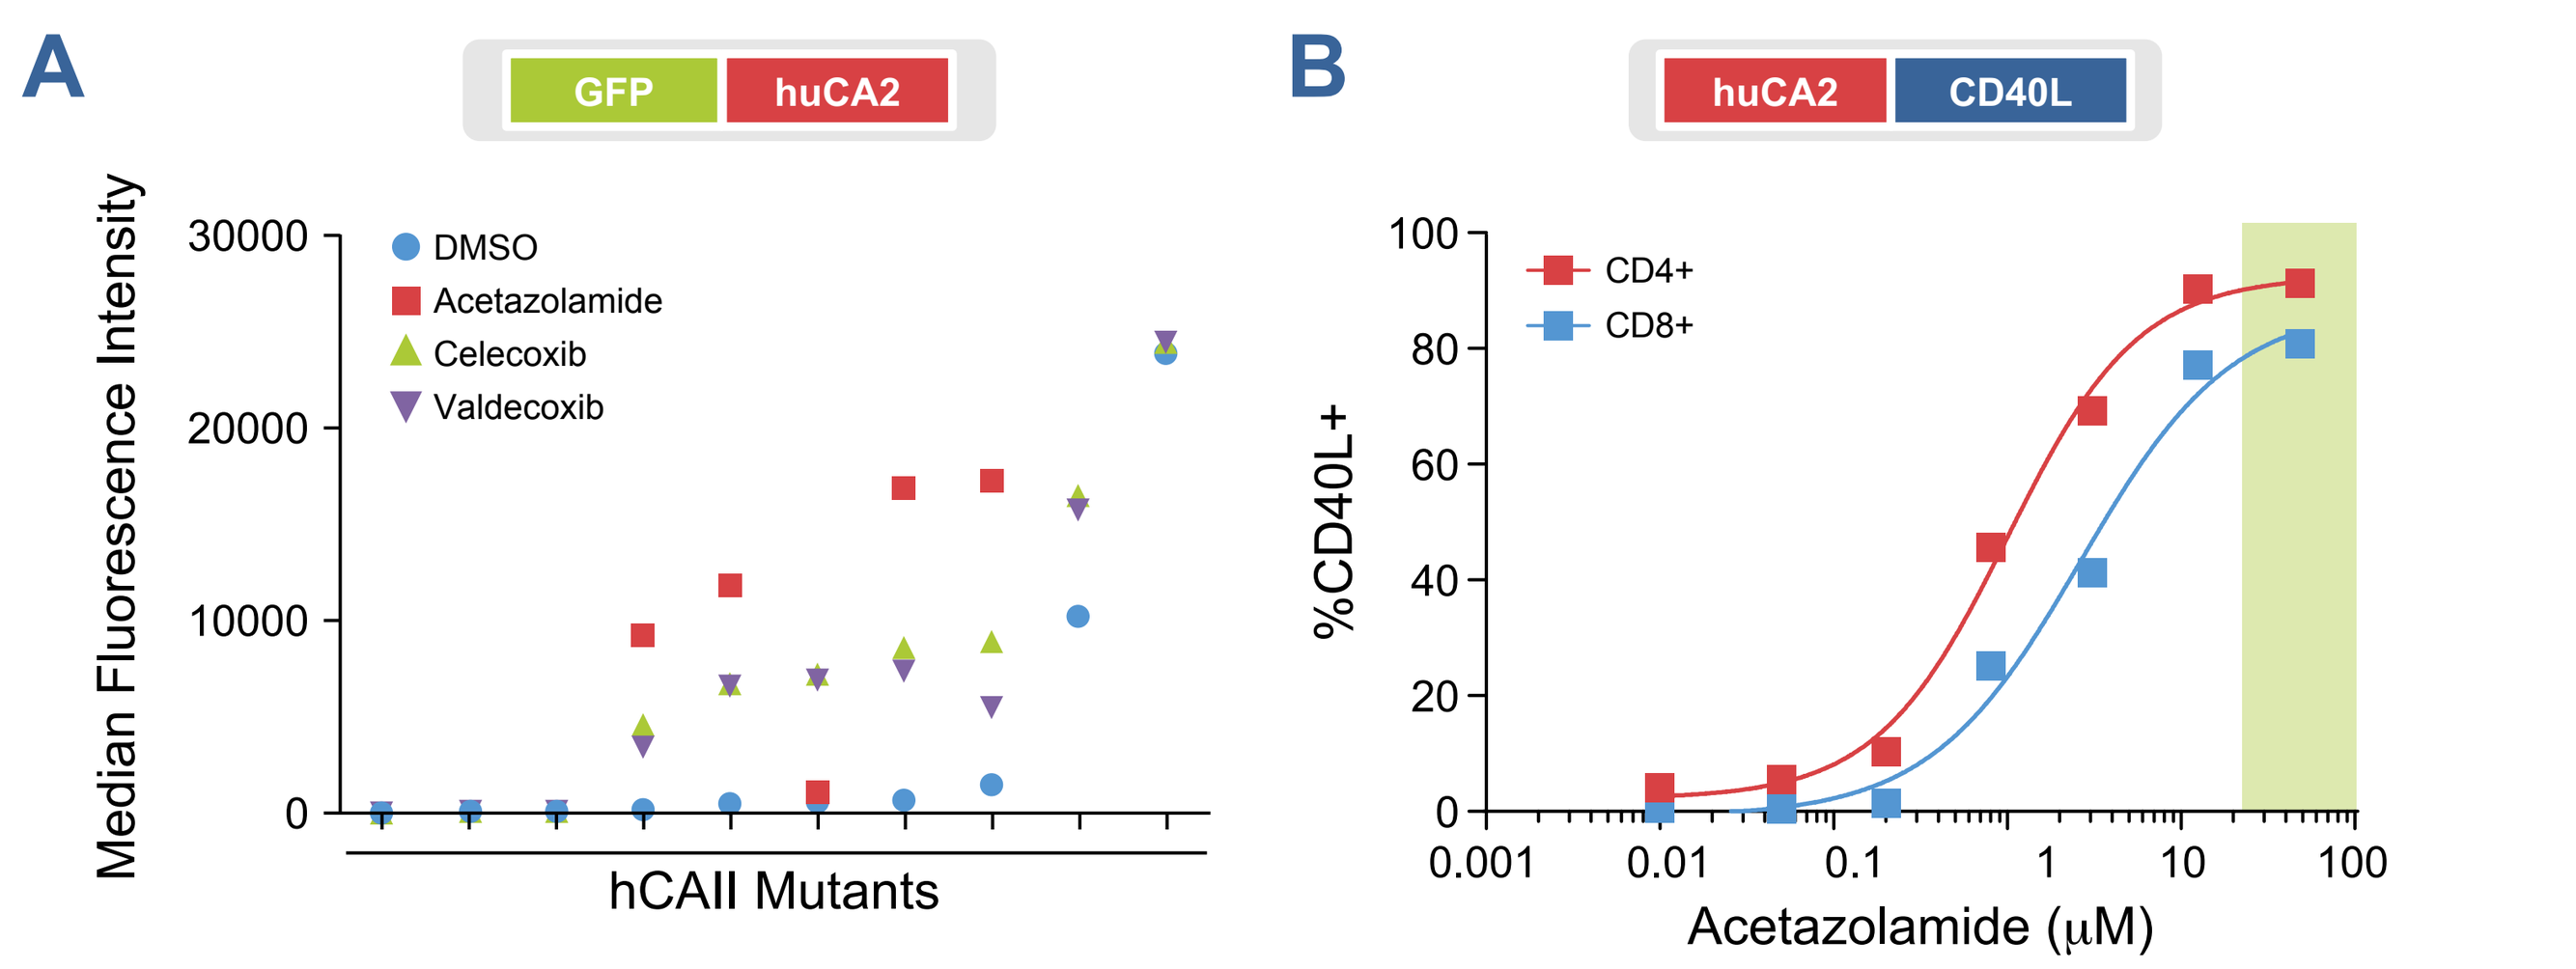
A
GFP
huCA2
Median Fluorescence Intensity
DMSO
Acetazolamide
Celecoxib
Valdecoxib
hCAII Mutants
B
huCA2
CD40L
%CD40L+
CD4+
CD8+
Acetazolamide (μM)

A) A mutagenesis library of the human carbonic anhydrase 2 (huCA2) protein domain was fused to GFP and subjected to a DD selection campaign. Individual clones from selected pools were screened for DD activity in the absence or presence of the stabilizing ligand, acetazolamide, as measured by flow cytometry. **B**) Activated T cells were transduced with a CA2-regulated CD40L construct. Dose response curve of acetazolamide regulating surface levels of CA2-CD40L are plotted as gated percent positive. The green area is an approximate range of acetazolamide Cmax levels in the plasma of humans from various clinical studies.

Figure 6: IL12 Improves Expansion and Efficacy of CD19 CAR *in vivo*

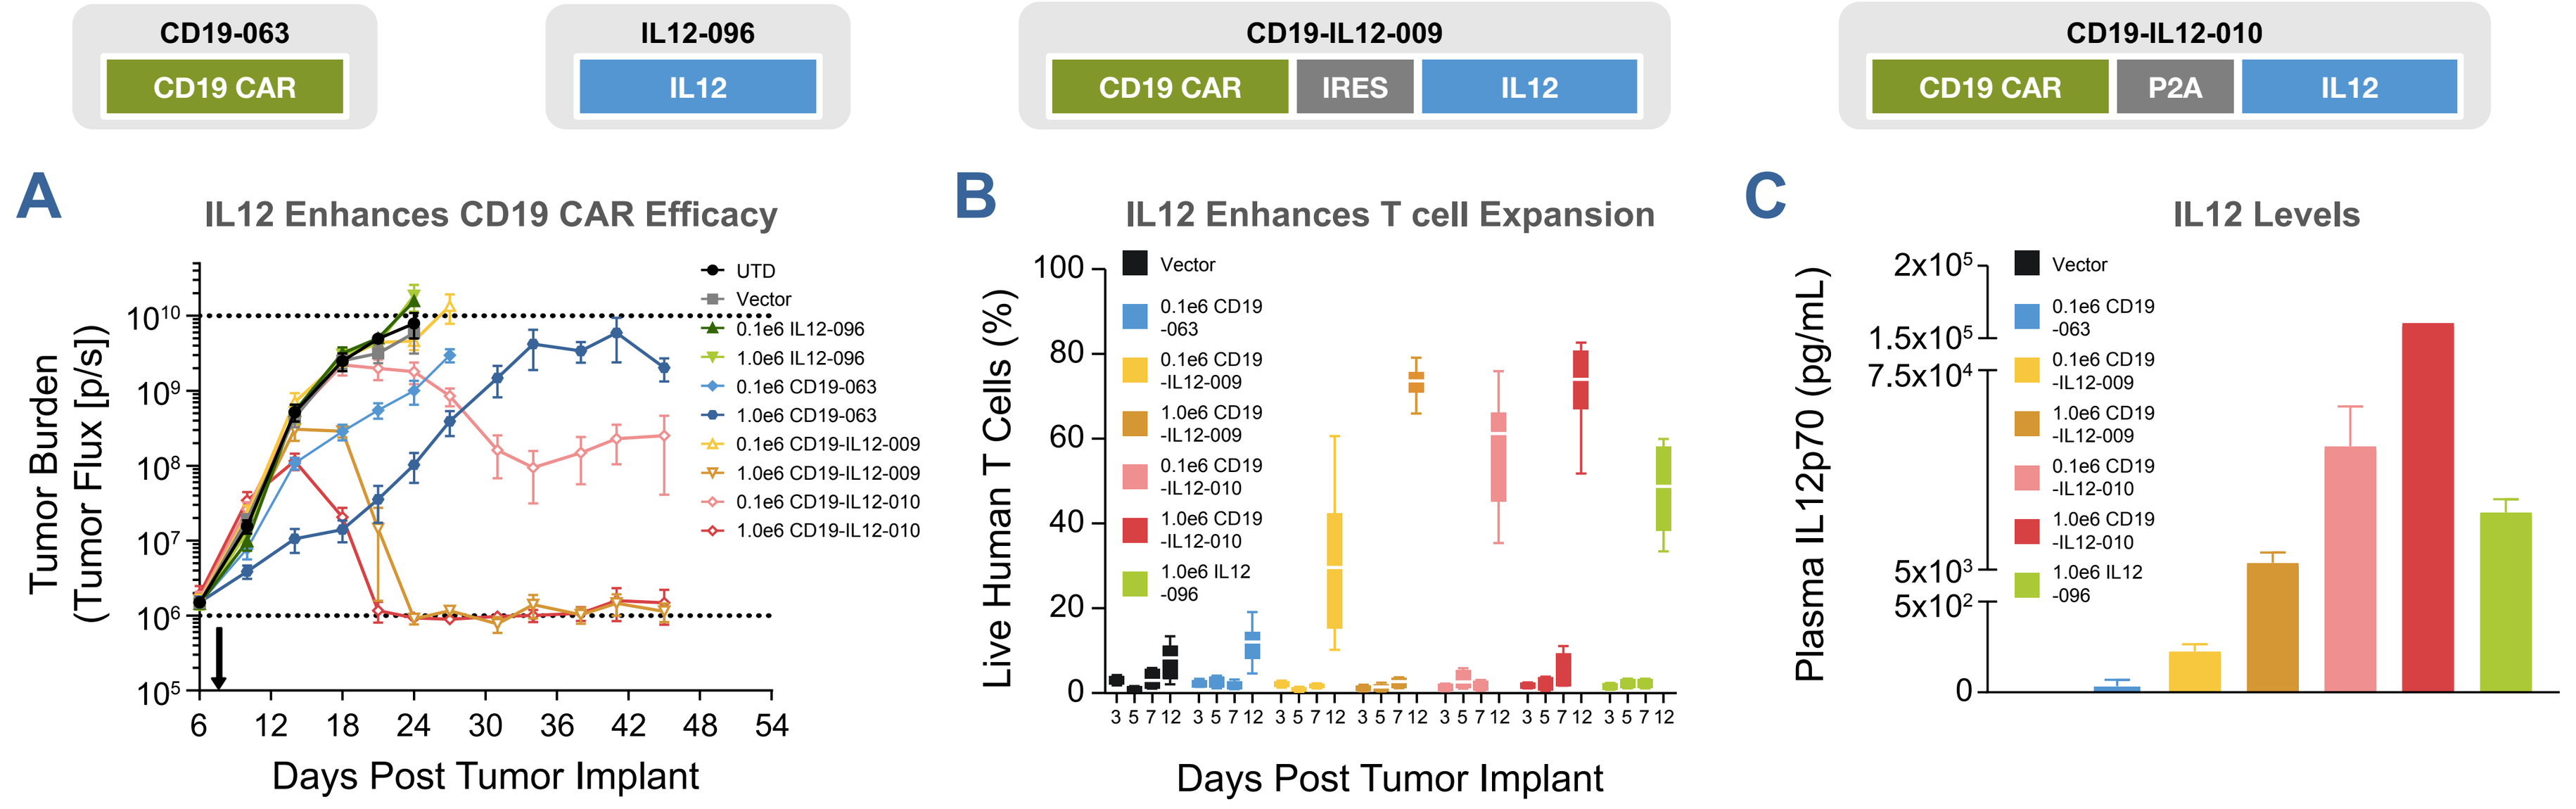
CD19-063
CD19 CAR
IL12-096
IL12
CD19-IL12-009
CD19 CAR
IRES
IL12
CD19-IL12-010
CD19 CAR
P2A
IL12
A
IL12 Enhances CD19 CAR Efficacy
Tumor Burden (Tumor Flux [p/s])
Days Post Tumor Implant
B
IL12 Enhances T cell Expansion
Live Human T Cells (%)
Days Post Tumor Implant
C
IL12 Levels
Plasma IL12p70 (pg/mL)

A) Nalm6-luc tumor-bearing NSG mice were injected with CAR-T cells at 10^5 or 10^6 cells/mouse 7 days post tumor implantation (n = 8/group). Tumor burden was quantified using bioluminescence upon luciferin administration. **B**) Circulating human T cells were quantified using CD45 as a FACS marker and (**C**) IL12 levels quantified from plasma on day 12 using an MSD assay.

Figure 7: Drug-dependent Regulation of IL12 in CAR-T Cells *in vitro* and *in vivo* with a Clinically Translatable DD

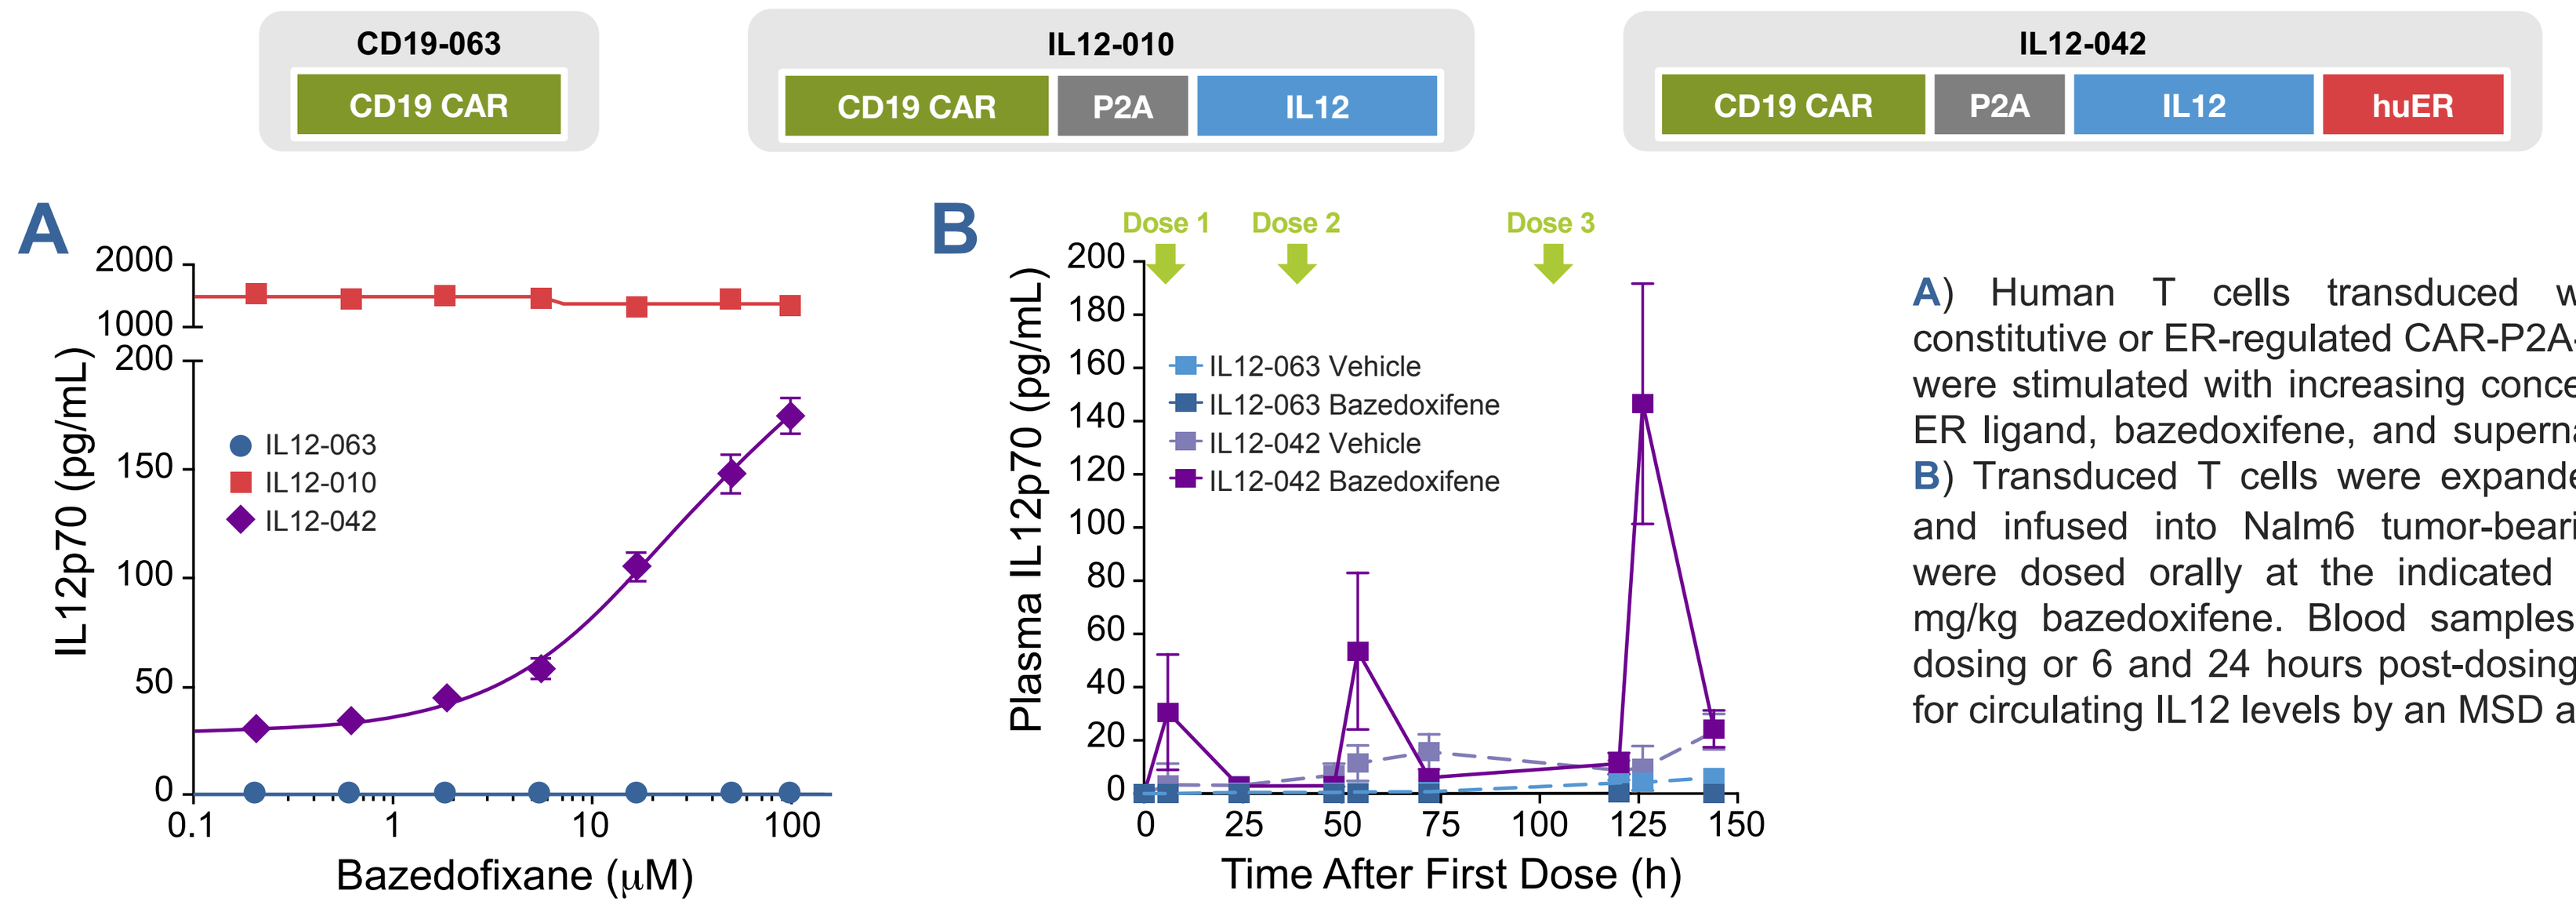
CD19-063
CD19 CAR
IL12-010
CD19 CAR
P2A
IL12
IL12-042
CD19 CAR
P2A
IL12
huER
A
IL12p70 (pg/mL)
IL12-063
IL12-010
IL12-042
Bazedofixane (μM)
B
Dose 1
Dose 2
Dose 3
Plasma IL12p70 (pg/mL)
IL12-063 Vehicle
IL12-063 Bazedoxifene
IL12-042 Vehicle
IL12-042 Bazedoxifene
Time After First Dose (h)

A) Human T cells transduced with either the constitutive or ER-regulated CAR-P2A-IL12 constructs were stimulated with increasing concentrations of the ER ligand, bazedoxifene, and supernatants assayed. **B**) Transduced T cells were expanded for 10 days, and infused into Nalm6 tumor-bearing mice. Mice were dosed orally at the indicated times with 100 mg/kg bazedoxifene. Blood samples taken prior to dosing or 6 and 24 hours post-dosing were analyzed for circulating IL12 levels by an MSD assay.

Figure 8: Expression and Regulation of a Membrane-bound Form of IL12 on T Cells

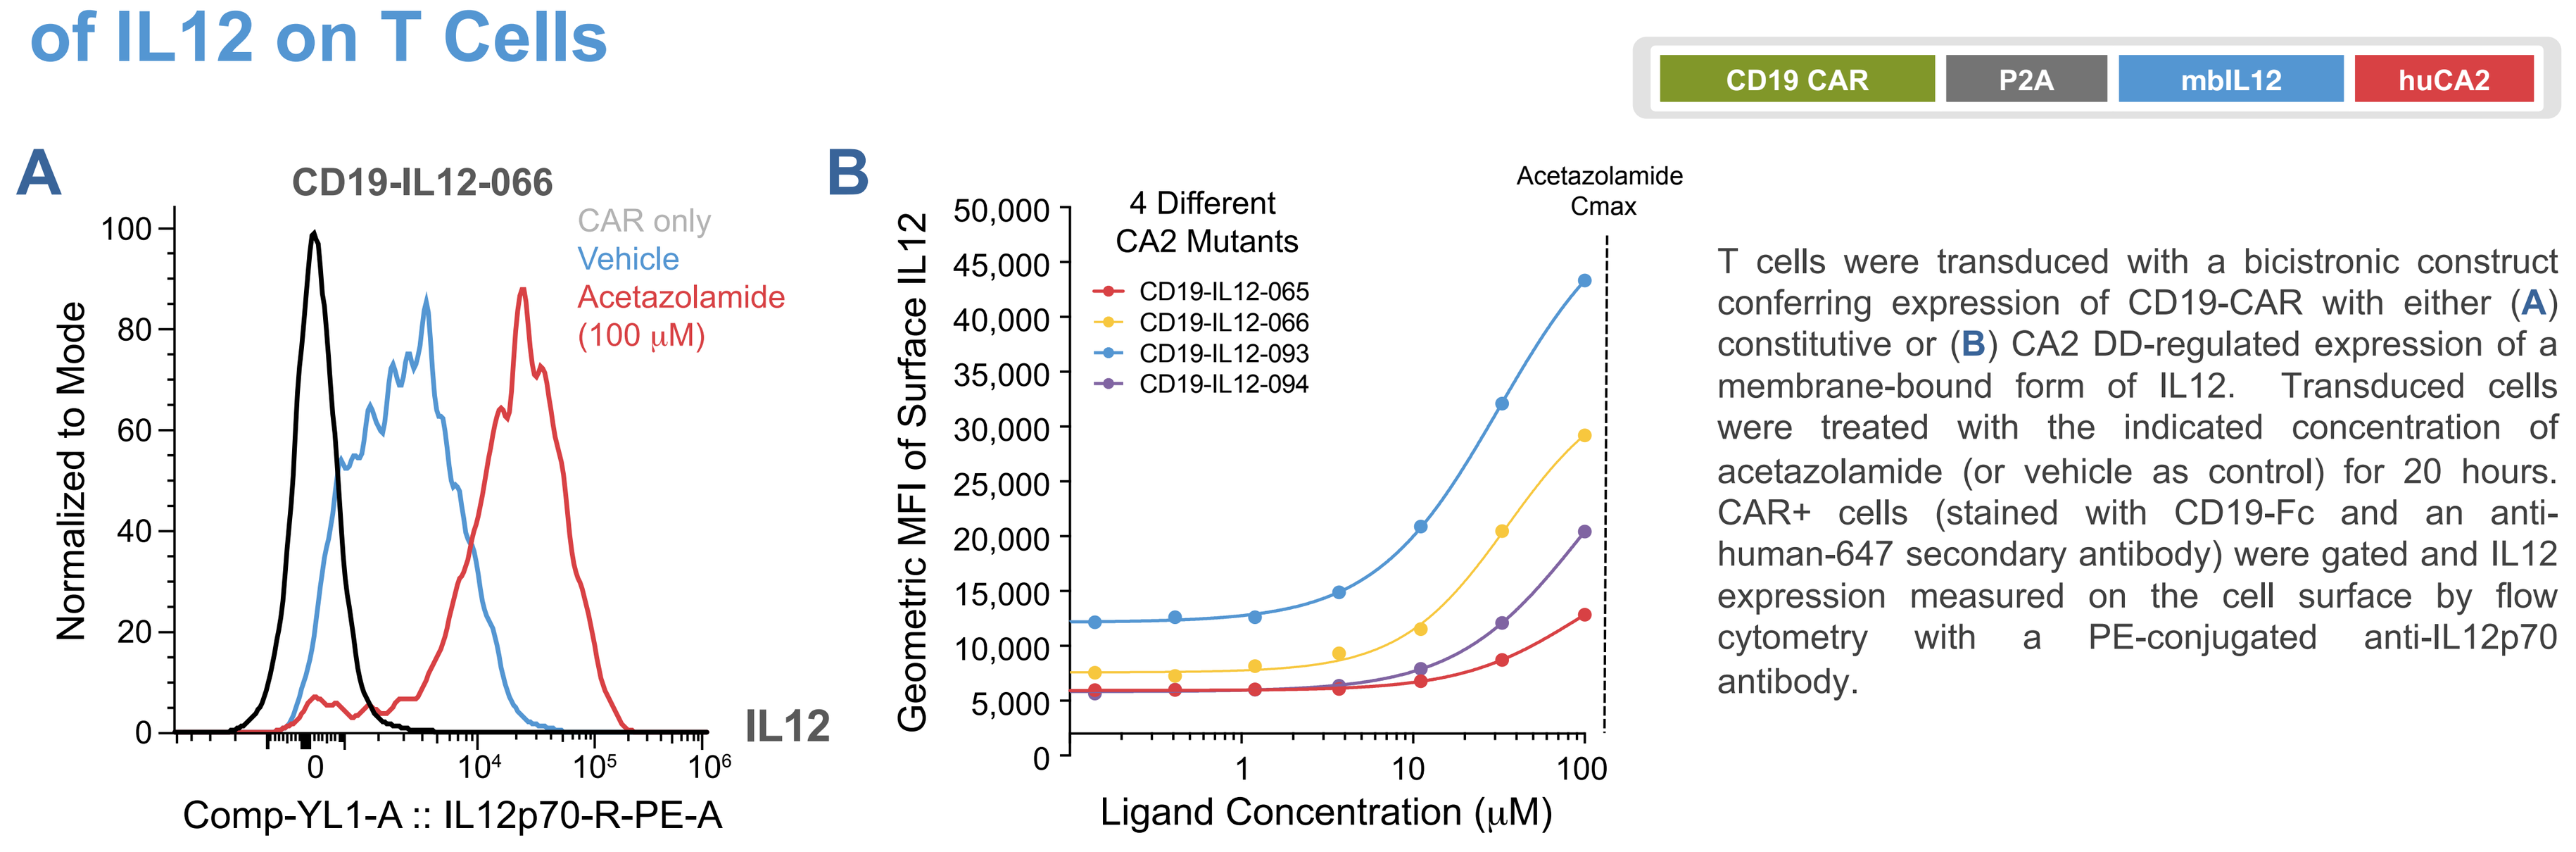
CD19 CAR
P2A
mbIL12
huCA2
A
CD19-IL12-066
CAR only
Vehicle
Acetazolamide (100 μM)
Normalized to Mode
IL12
Comp-YL1-A :: IL12p70-R-PE-A
B
4 Different CA2 Mutants
CD19-IL12-065
CD19-IL12-066
CD19-IL12-093
CD19-IL12-094
Acetazolamide Cmax
Geometric MFI of Surface IL12
Ligand Concentration (μM)

T cells were transduced with a bicistronic construct conferring expression of CD19-CAR with either (**A**) constitutive or (**B**) CA2 DD-regulated expression of a membrane-bound form of IL12. Transduced cells were treated with the indicated concentration of acetazolamide (or vehicle as control) for 20 hours. CAR+ cells (stained with CD19-Fc and an anti-human-647 secondary antibody) were gated and IL12 expression measured on the cell surface by flow cytometry with a PE-conjugated anti-IL12p70 antibody.

SUMMARY

- Obsidian's Destabilizing Domain (DD) technology provides titratable and reversible regulation of protein levels via exogenous small molecule dosing.
- Multiple human DD families have been created, each of which can be regulated by distinct FDA approved drugs.
- We have demonstrated titratable small molecule regulation of both CD40L and IL12 expression and activity in human T cells *in vitro* and *in vivo*.
- With precise control over levels of immunomodulatory factors produced, we are building next generation CAR-T cell products for enhanced efficacy against solid tumors and more favorable safety profiles.

OBSIDIAN™
Obsidian Therapeutics, Inc. - 1030 Massachusetts Avenue – Cambridge, MA 02138 - info@obsidiantx.com