Poster #444

Feasibility of using core needle biopsy technique for minimally invasive tumor procurement to generate OBX-115 TIL from NSCLC tumors

Matthew Bott[1]*; Zheng Ao[2]*; Alonso Villasmil Ocando[2]; Nirzari Shah[2]; Bulent Arman Aksoy[2]; James Isaac Godfroy[2]; Rachel Burga[2]; Kathryn Aron[2]; Sheyla Mirabal[2]; Tyler Humpton[2]; Vy Le[2]; Gauri Kulkarni[2]; Ngoc Ly[2]; Giridharan Ramsingh[2]; Gwendolyn Wilmes[2]; Chirag Khatiwala[2]; Jan ter Meulen[2]; Anand Veerapathran[2]; Michelle Ols[2]; Phoebe Clark[1]; Sophie Hieronymi[1]; Stephen B Solomon[1]; Adam J Schoenfeld[1]

1. Memorial Sloan Kettering Cancer Center, New York, NY, USA. 2. Obsidian Therapeutics Inc., Cambridge, MA, USA.

zao@obsidiantx.com

Introduction

- Conventional non-engineered tumor-infiltrating lymphocyte (TIL) cell therapy has demonstrated antitumor activity in metastatic non-small cell lung cancer (NSCLC)[1]
- Historically, TIL for cell therapy are manufactured from surgically excised tumors; however, eligibility for surgery may be a barrier to treatment for many patients with NSCLC
- cytoTIL15™ cells (OBX-115) are engineered with membrane-bound IL15 (mbIL15) fused to a carbonic anhydrase 2 (CA2) drug-responsive domain (DRD) to enable regulation using the FDA-approved small-molecule ligand acetazolamide (ACZ), and have demonstrated IL2-independent expansion and preclinical functionality using multiple solid tumor types, including NSCLC[2]
- In a first-in-human phase 1 study, OBX-115 demonstrated favorable drug-product attributes (CD8+ enriched, minimally exhausted, "stem-like" progenitor T-cell phenotype) and produced an objective response rate (ORR) of 44% in 9 efficacy-evaluable patients with advanced melanoma[3]
- We hypothesized that the unique OBX-115 manufacturing process would require less tumor tissue to generate NSCLC TIL, without compromising product characteristics, based on our past successful experience manufacturing from clinical core needle biopsies (cCNB) from melanoma[4]
- To test this hypothesis, tumor samples were collected in pairs from surgically resected tissue, sCNB performed on the same tumor tissue post-excision, and cCNB separately (**Figure 1**), from which we generated OBX-115 and non-engineered TIL. TIL phenotype as well as T-cell receptor (TCR) repertoires were evaluated via flow cytometry and TCR-seq respectively. TIL cytotoxicity was evaluated in 3D tumor-spheroid models generated from autologous patient-derived tumor cell lines (PDc)

Figure 1. CNB tumor tissue is simulated from paired surgical excision

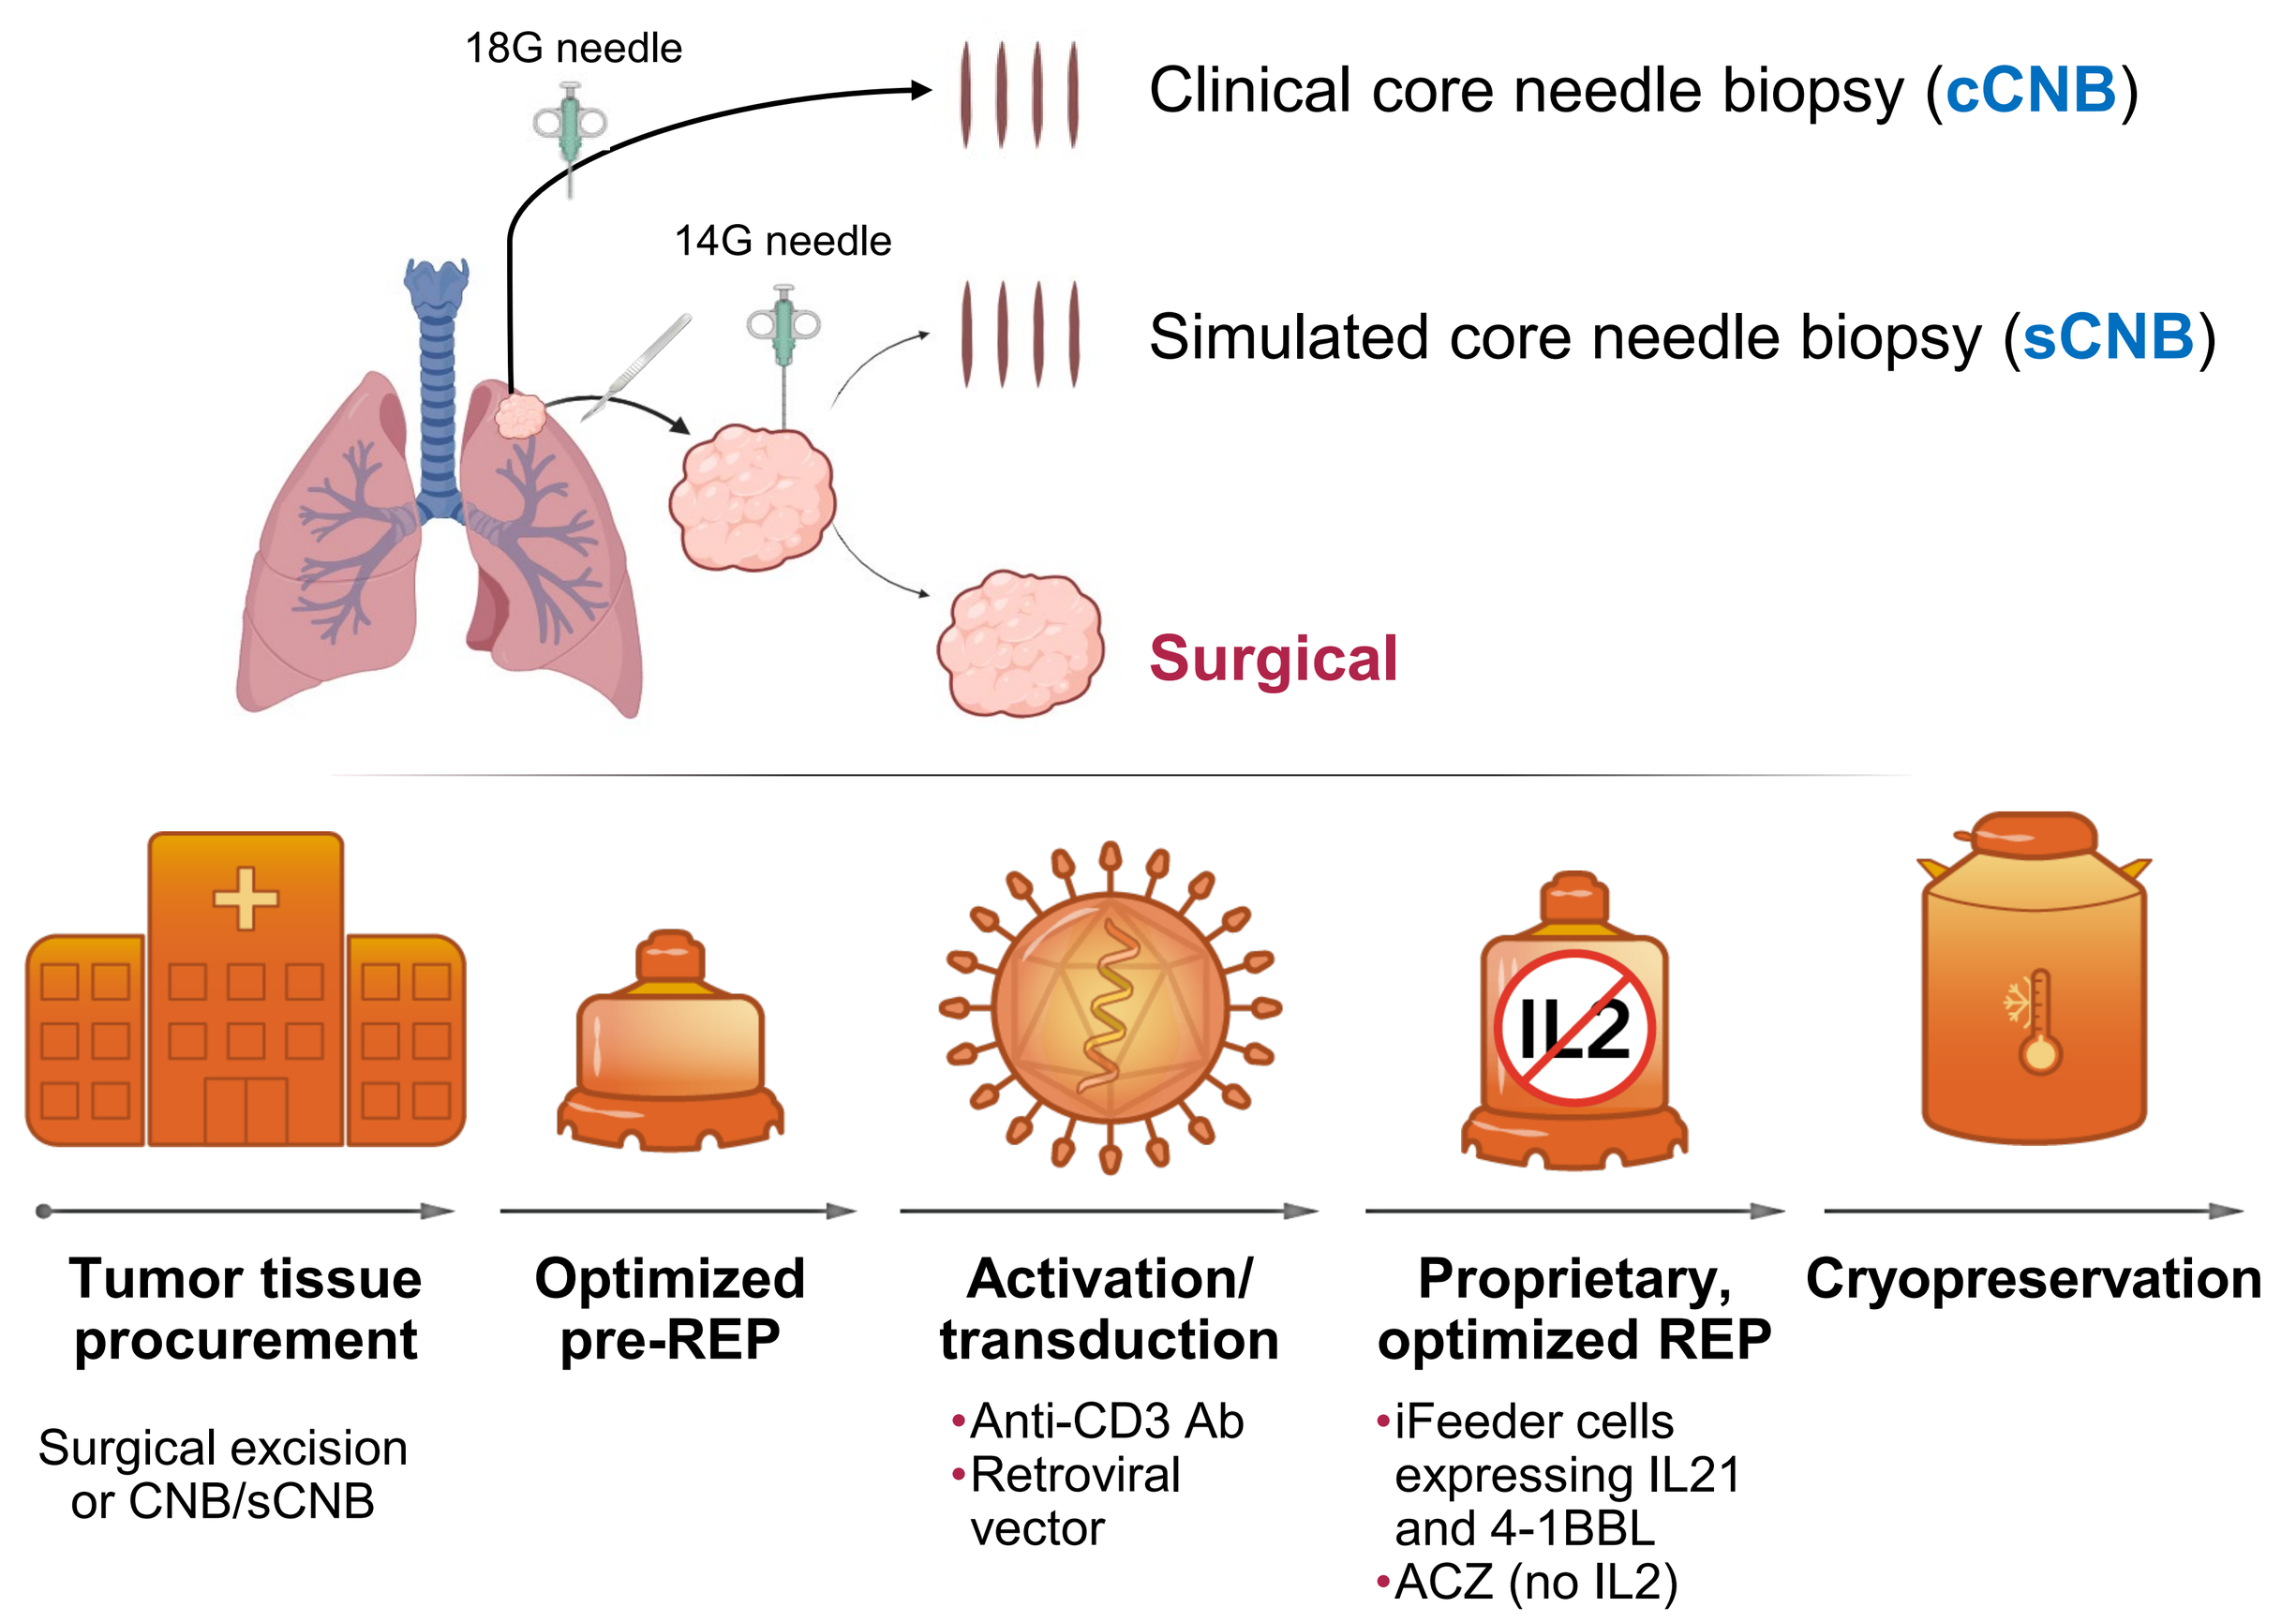
18G needle
Clinical core needle biopsy (cCNB)
14G needle
Simulated core needle biopsy (sCNB)
Surgical
IL2
Tumor tissue procurement
Surgical excision or CNB/sCNB
Optimized pre-REP
Activation/ transduction
•Anti-CD3 Ab
•Retroviral vector
Proprietary, optimized REP
•iFeeder cells expressing IL21 and 4-1BBL
•ACZ (no IL2)
Cryopreservation

Figure 1. Generation of OBX-115 TIL from NSCLC using paired surgical excision and sCNB or cCNB tumor tissue.

Results

Figure 2. Representative images of processed surgical, sCNB, and cCNB tumor tissue

Donor 3 Surgical	Donor 3 sCNB	Donor 5 cCNB

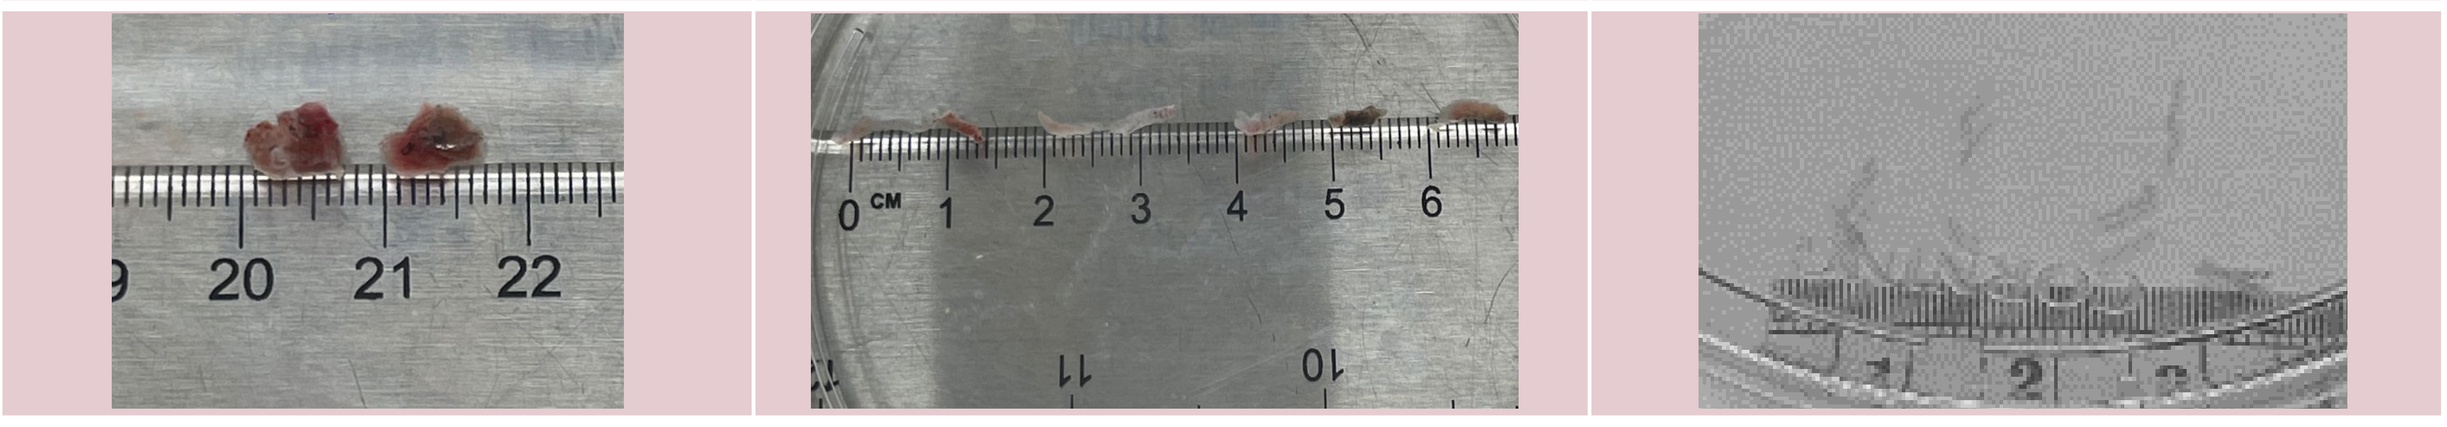

Figure 2. Representative images of processed surgical, sCNB, and cCNB tumor tissue.

Results (*continued*)

Figure 3. Similar pre-REP TIL yield from surgical and sCNB tumor tissue

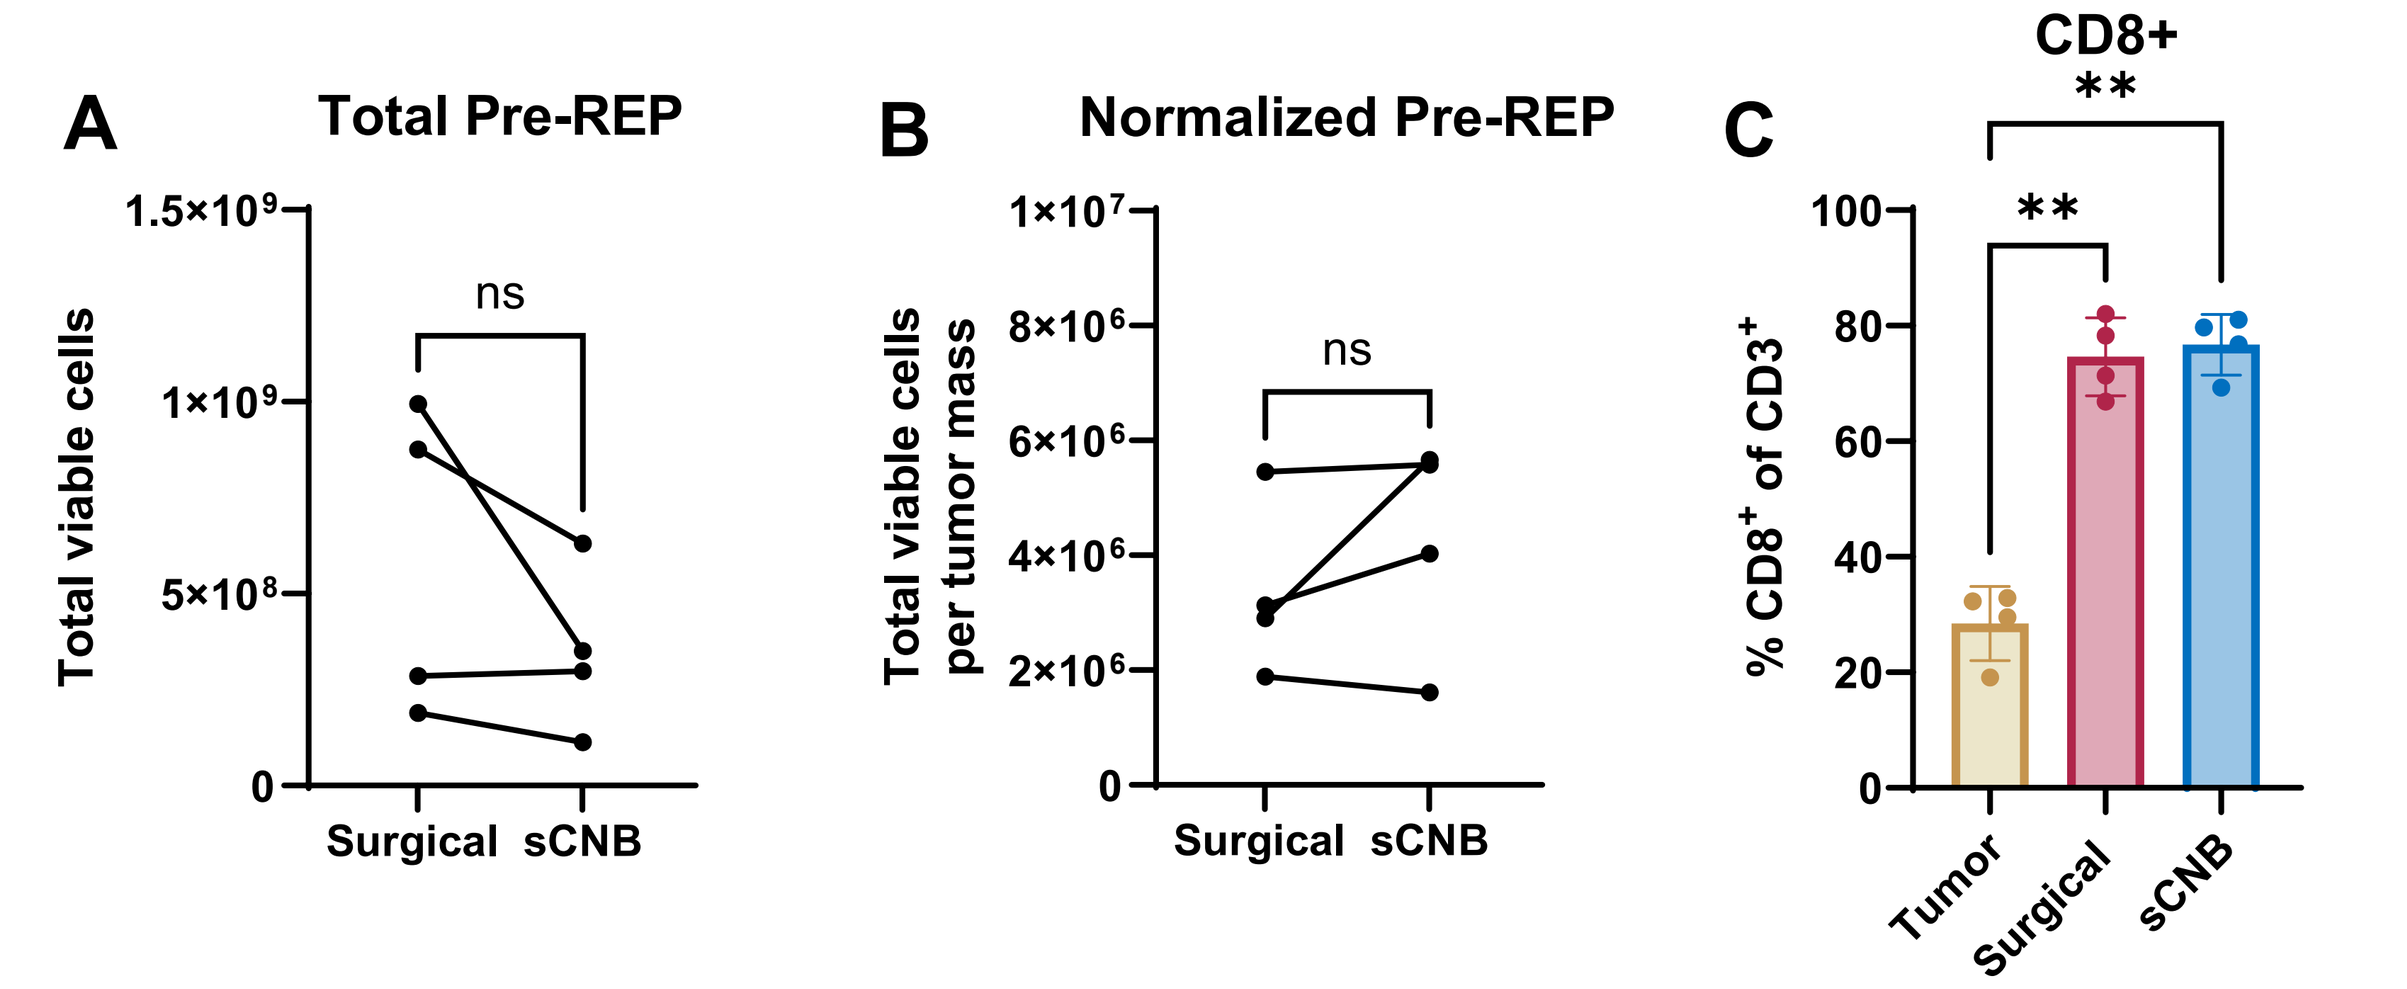
A
Total Pre-REP
Total viable cells
Surgical sCNB
ns
B
Normalized Pre-REP
Total viable cells per tumor mass
C
CD8+
% CD8+ of CD3+
Tumor Surgical sCNB
**

Figure 3. Surgical and sCNB tumor tissue yielded similar amounts of pre-REP TIL. Paired pre-REP expansion of surgical resection and sCNB led to similar total pre-REP cell yield regardless of starting tumor mass (**A–B**, student's T test, p<0.05). Pre-REP TIL expansion with tumor tissue from either surgical excision or sCNB led to similar enrichment of CD8+ T cells relative to starting CD8+ frequency in the tumor (**C**, n=4 **p<0.01, one-way ANOVA with Tukey's HSD).

Figure 4. Similar OBX-115 TIL generation from surgical and sCNB tumor tissue

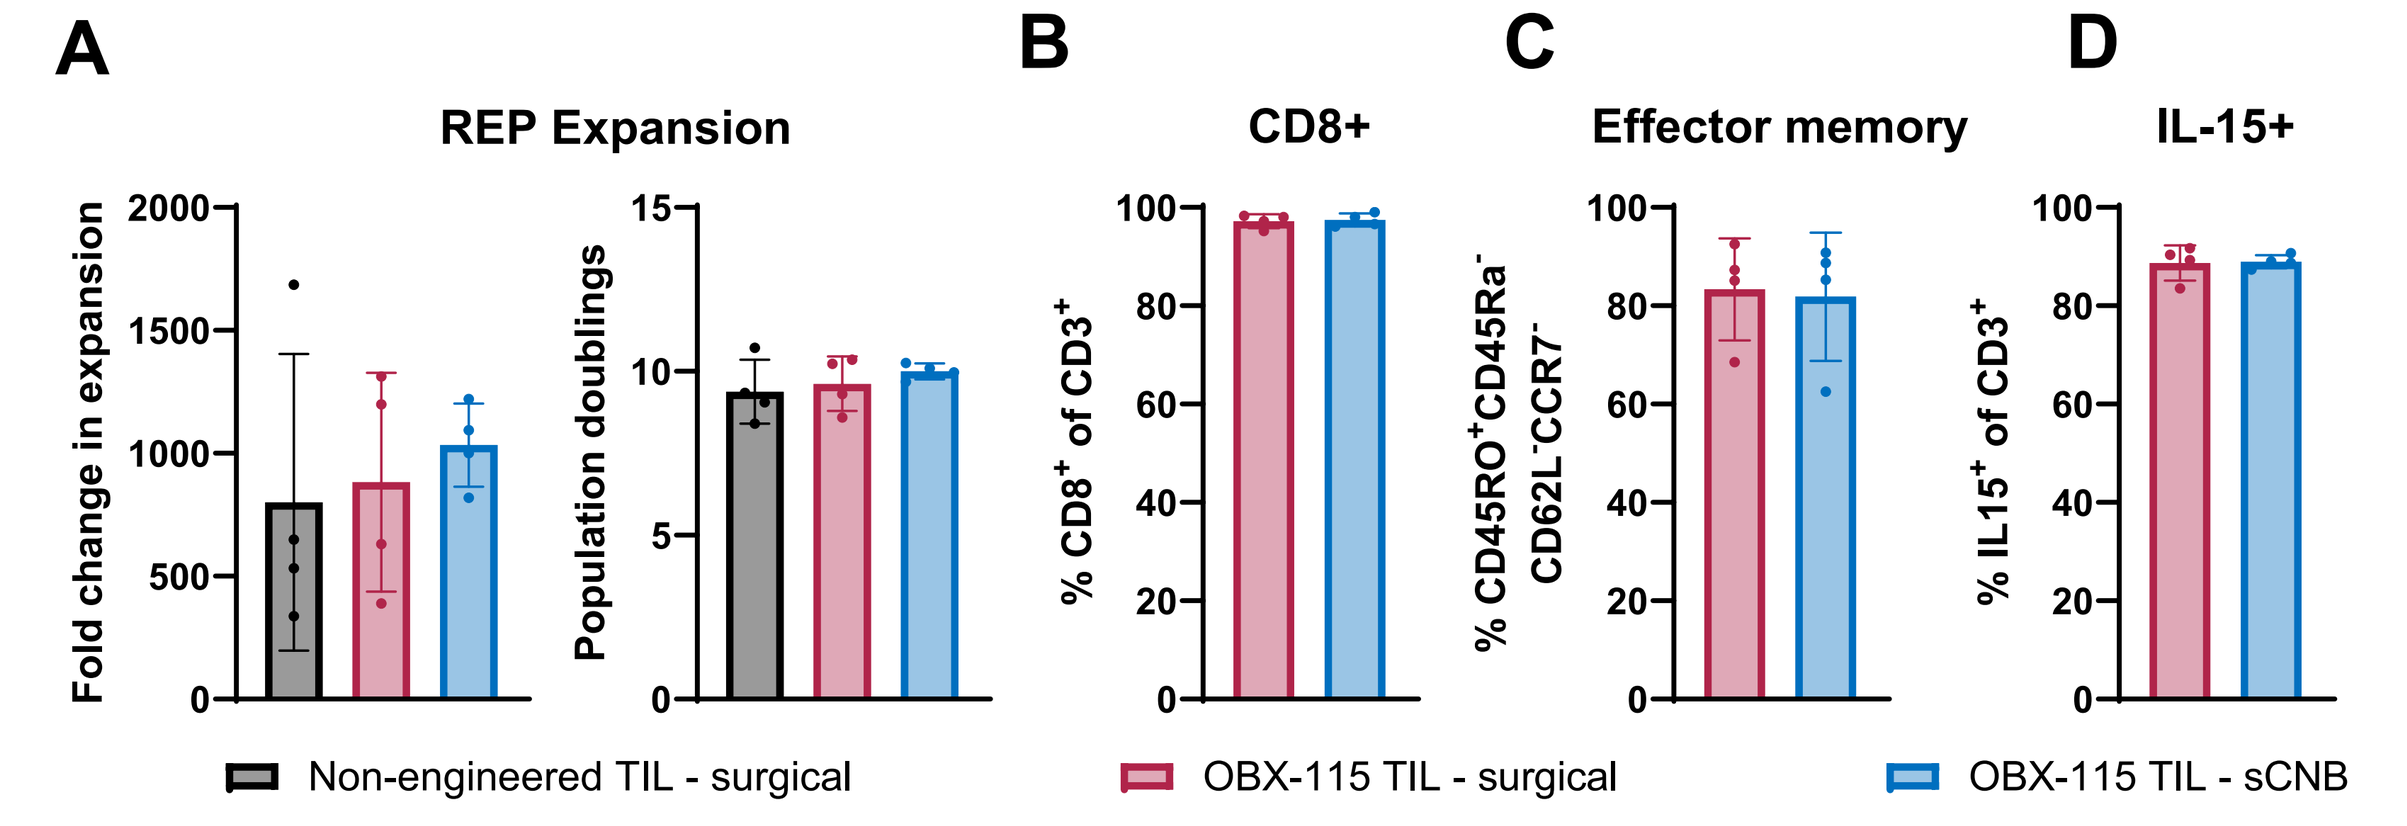
A
REP Expansion
Fold change in expansion
Population doublings
B
CD8+
% CD8+ of CD3+
C
Effector memory
% CD45RO+CD45Ra- CD62L-CCR7-
D
IL-15+
% IL15+ of CD3+
Non-engineered TIL - surgical
OBX-115 TIL - surgical
OBX-115 TIL - sCNB

Figure 4. OBX-115 manufacturing is successful using surgical excision or sCNB. Pre-REP TIL from surgical excision or sCNB led to similar IL2-independent expansion of OBX-115 TIL, as well as IL2-dependent expansion of non-engineered TIL during REP (n=4) (**A**). OBX-115 TIL from surgical excision and sCNB were similarly enriched for CD8+ T cells (**B**), of primarily effector-memory phenotype (**C**), with high expression of DRD-regulated mbIL15 (**D**).

Figure 5. Surgical and sCNB tumor tissue yield TIL with similar TCR clonotypes

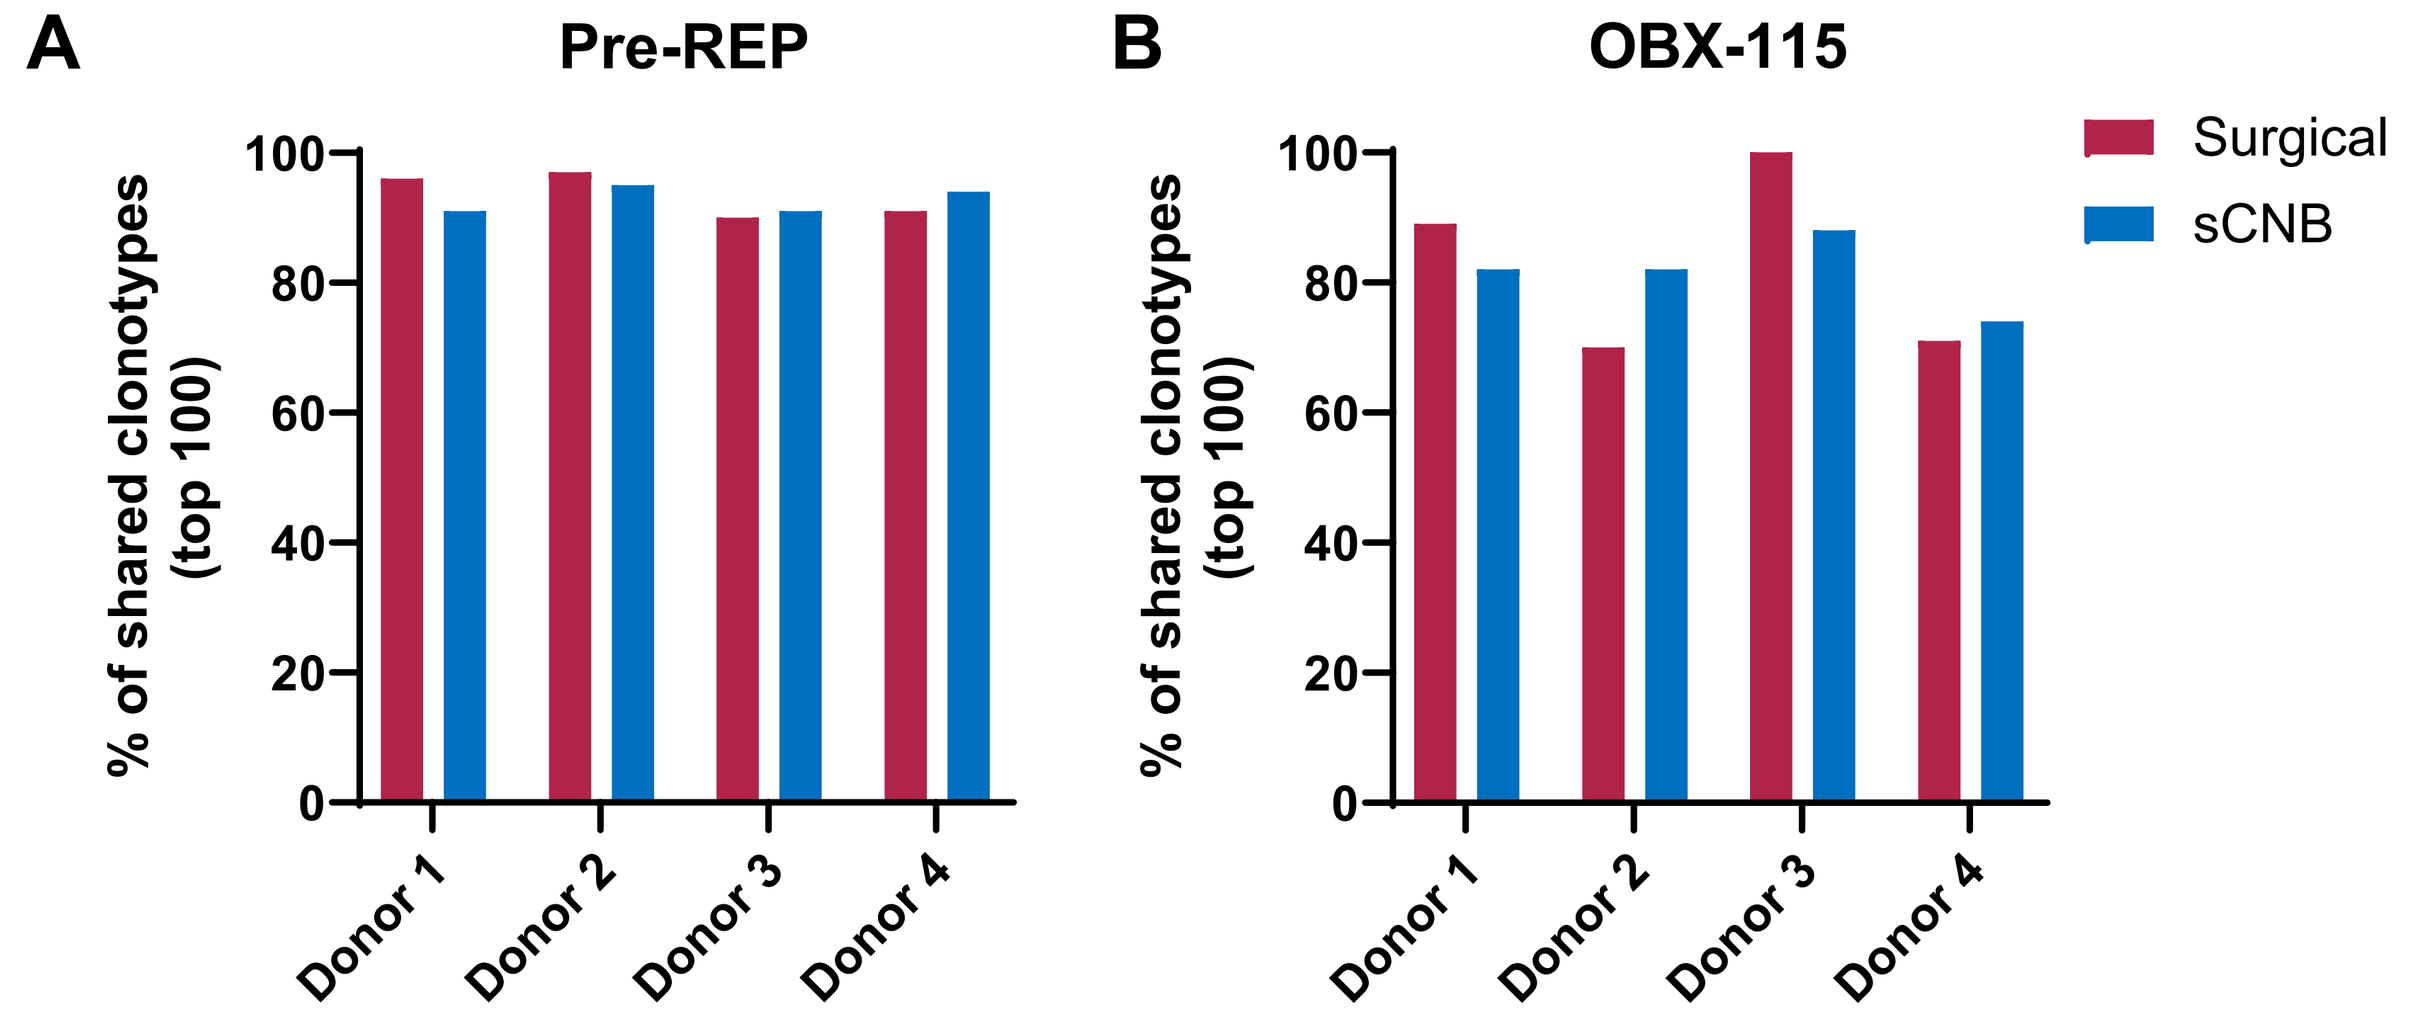
A
Pre-REP
B
OBX-115
% of shared clonotypes (top 100)
Donor 1
Donor 2
Donor 3
Donor 4
Surgical
sCNB

Figure 5. OBX-115 TIL generated from paired surgical and sCNB tissues show high similarity in their TCR profiles. For pairwise TCR repertoire comparison, the overlap of the top 100 clonotypes detected in one sample was calculated against its paired sample. Two-way comparison of top 100 clonotypes between paired surgical and sCNB tissue for pre-REP (**A**) and REP (**B**) TIL showed no statistical difference (pre-REP, p=0.70; REP, p=0.86; paired one-tail T-test).

Figure 6. OBX-115 TIL generated from both surgical/sCNB tissue had favorable phenotype compared with non-engineered TIL

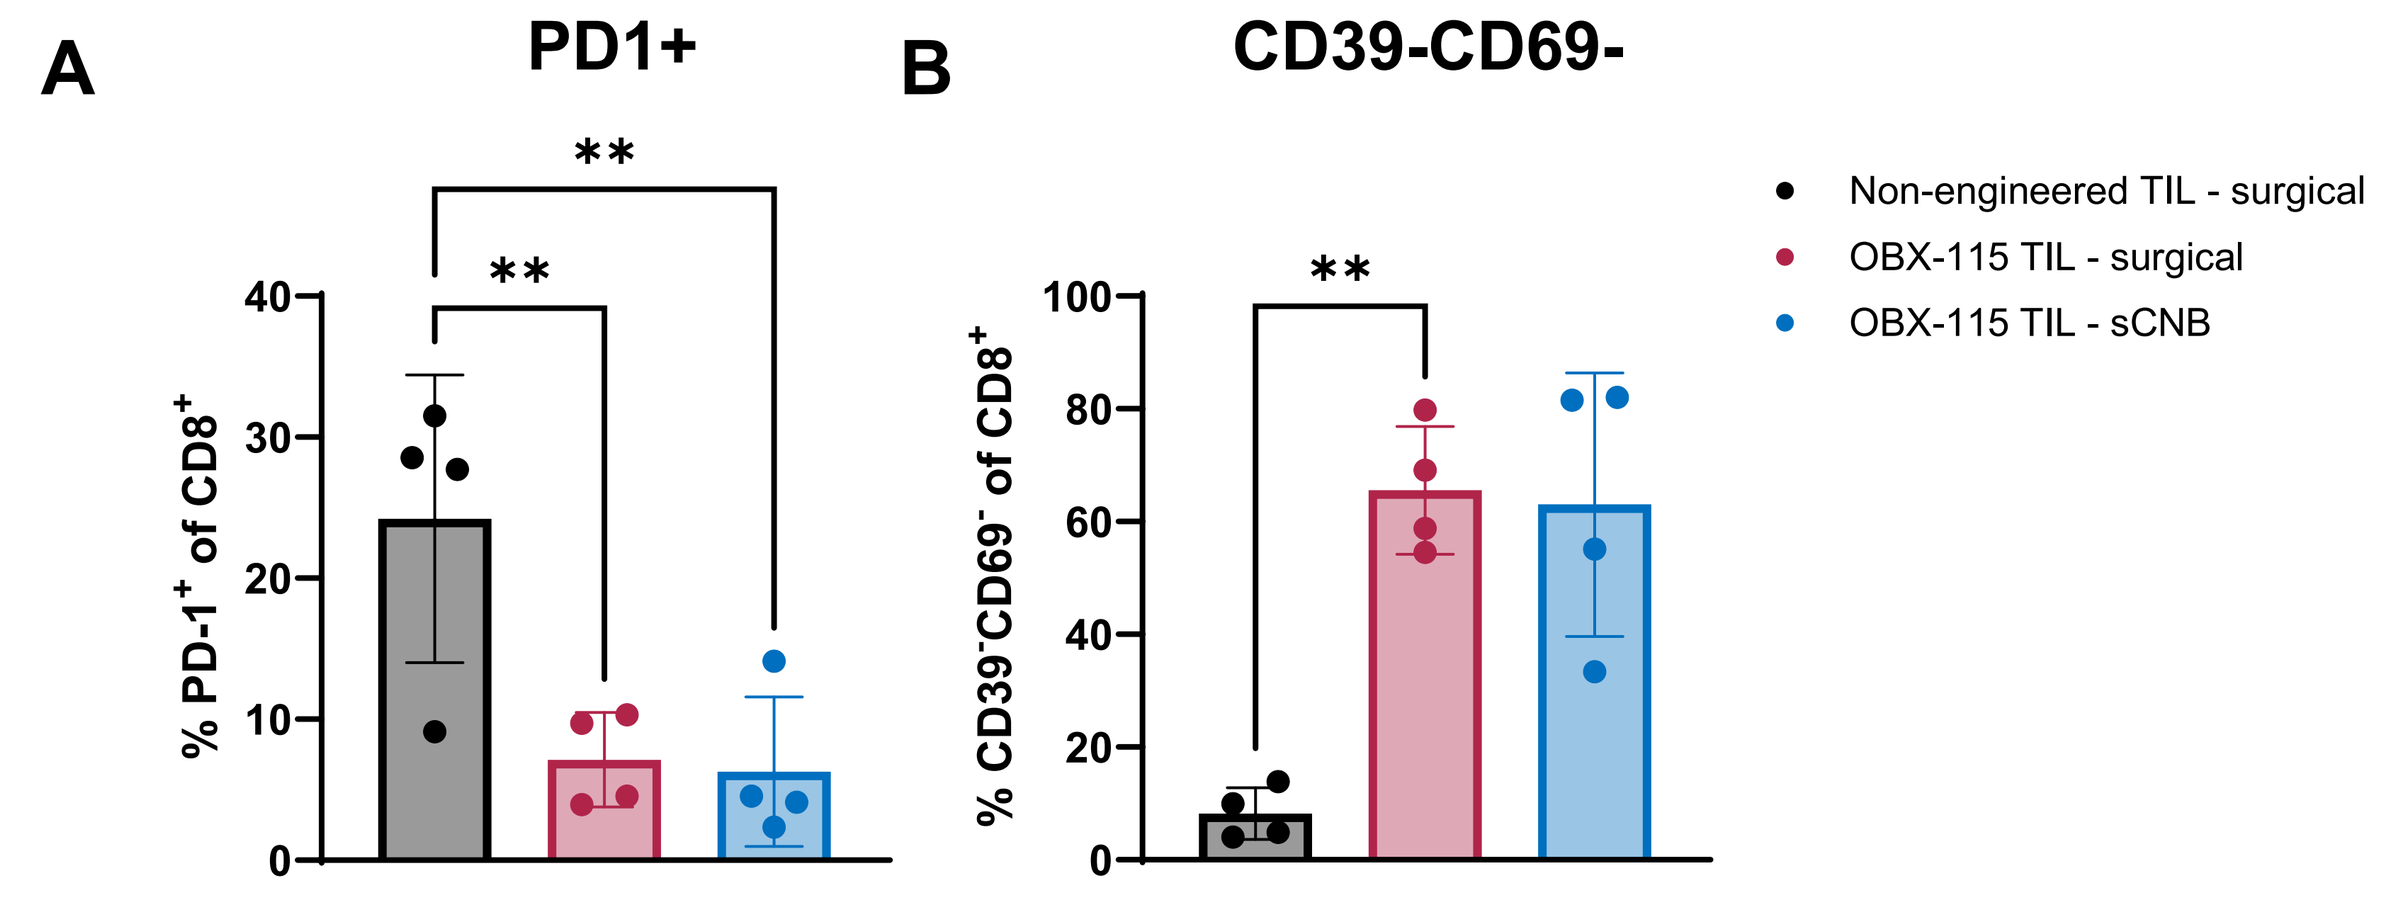
A
PD1+
% PD-1+ of CD8+
B
CD39-CD69-
% CD39-CD69- of CD8+
Non-engineered TIL - surgical
OBX-115 TIL - surgical
OBX-115 TIL - sCNB

Figure 6. OBX-115 TIL have lower proportions of PD-1+ and higher proportions of CD8+CD39-CD69- TIL. Research panel flow cytometry analysis revealed that at REP endpoint, OBX-115 TIL generated from surgical excision or sCNB had lower proportions of PD-1+ CD8+ T cells relative to non-engineered TIL (**A**). "Stem-like"[5] progenitor T cells, denoted by CD39 and CD69 downregulation, are enriched in OBX-115 TIL generated from surgical excision or sCNB tumor tissue relative to non-engineered TIL (**B**). (n=4, ** p< 0.01, * p<0.05; one-way ANOVA with Tukey's HSD). Non-engineered TIL are expanded in REP without IL15 engineering, high dose IL-2. and irradiated PBMC feeders.

Figure 7. Similar polyfunctionality of surgical and sCNB OBX-115 TIL

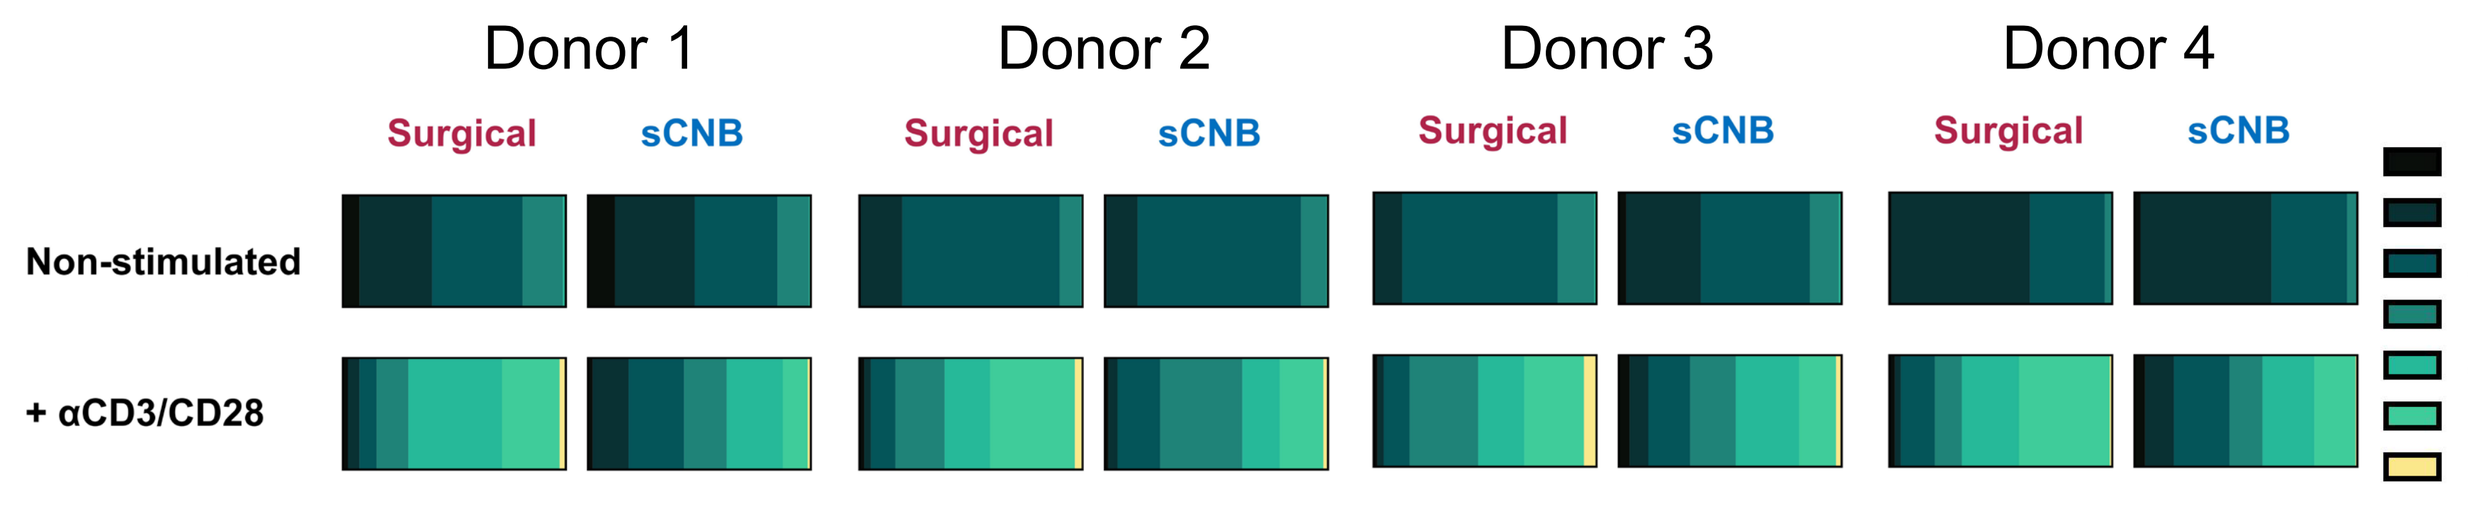
Donor 1
Donor 2
Donor 3
Donor 4
Surgical
sCNB
Non-stimulated
+ αCD3/CD28
0
1
2
3
4
5
6

Figure 7. OBX-115 TIL generated from paired surgical and sCNB tissues show similar polyfunctionality upon anti-CD3/anti-CD28 activation. In the presence of DRD-stabilizing ACZ, OBX-115 TIL generated from surgical excision and sCNB demonstrate similar polyfunctionality upon anti-CD3/anti-CD28 stimulation. Polyfunctionality is defined by frequency of cells co-expressing 0–6 of the following markers: CD107a, Granzyme B, Perforin, IFNγ, TNFα, IL2Rα, and IL2.

Figure 8. OBX-115 TIL generated from both surgical and sCNB tissue exert superior potent cytotoxicity against autologous tumor compared with non-engineered TIL

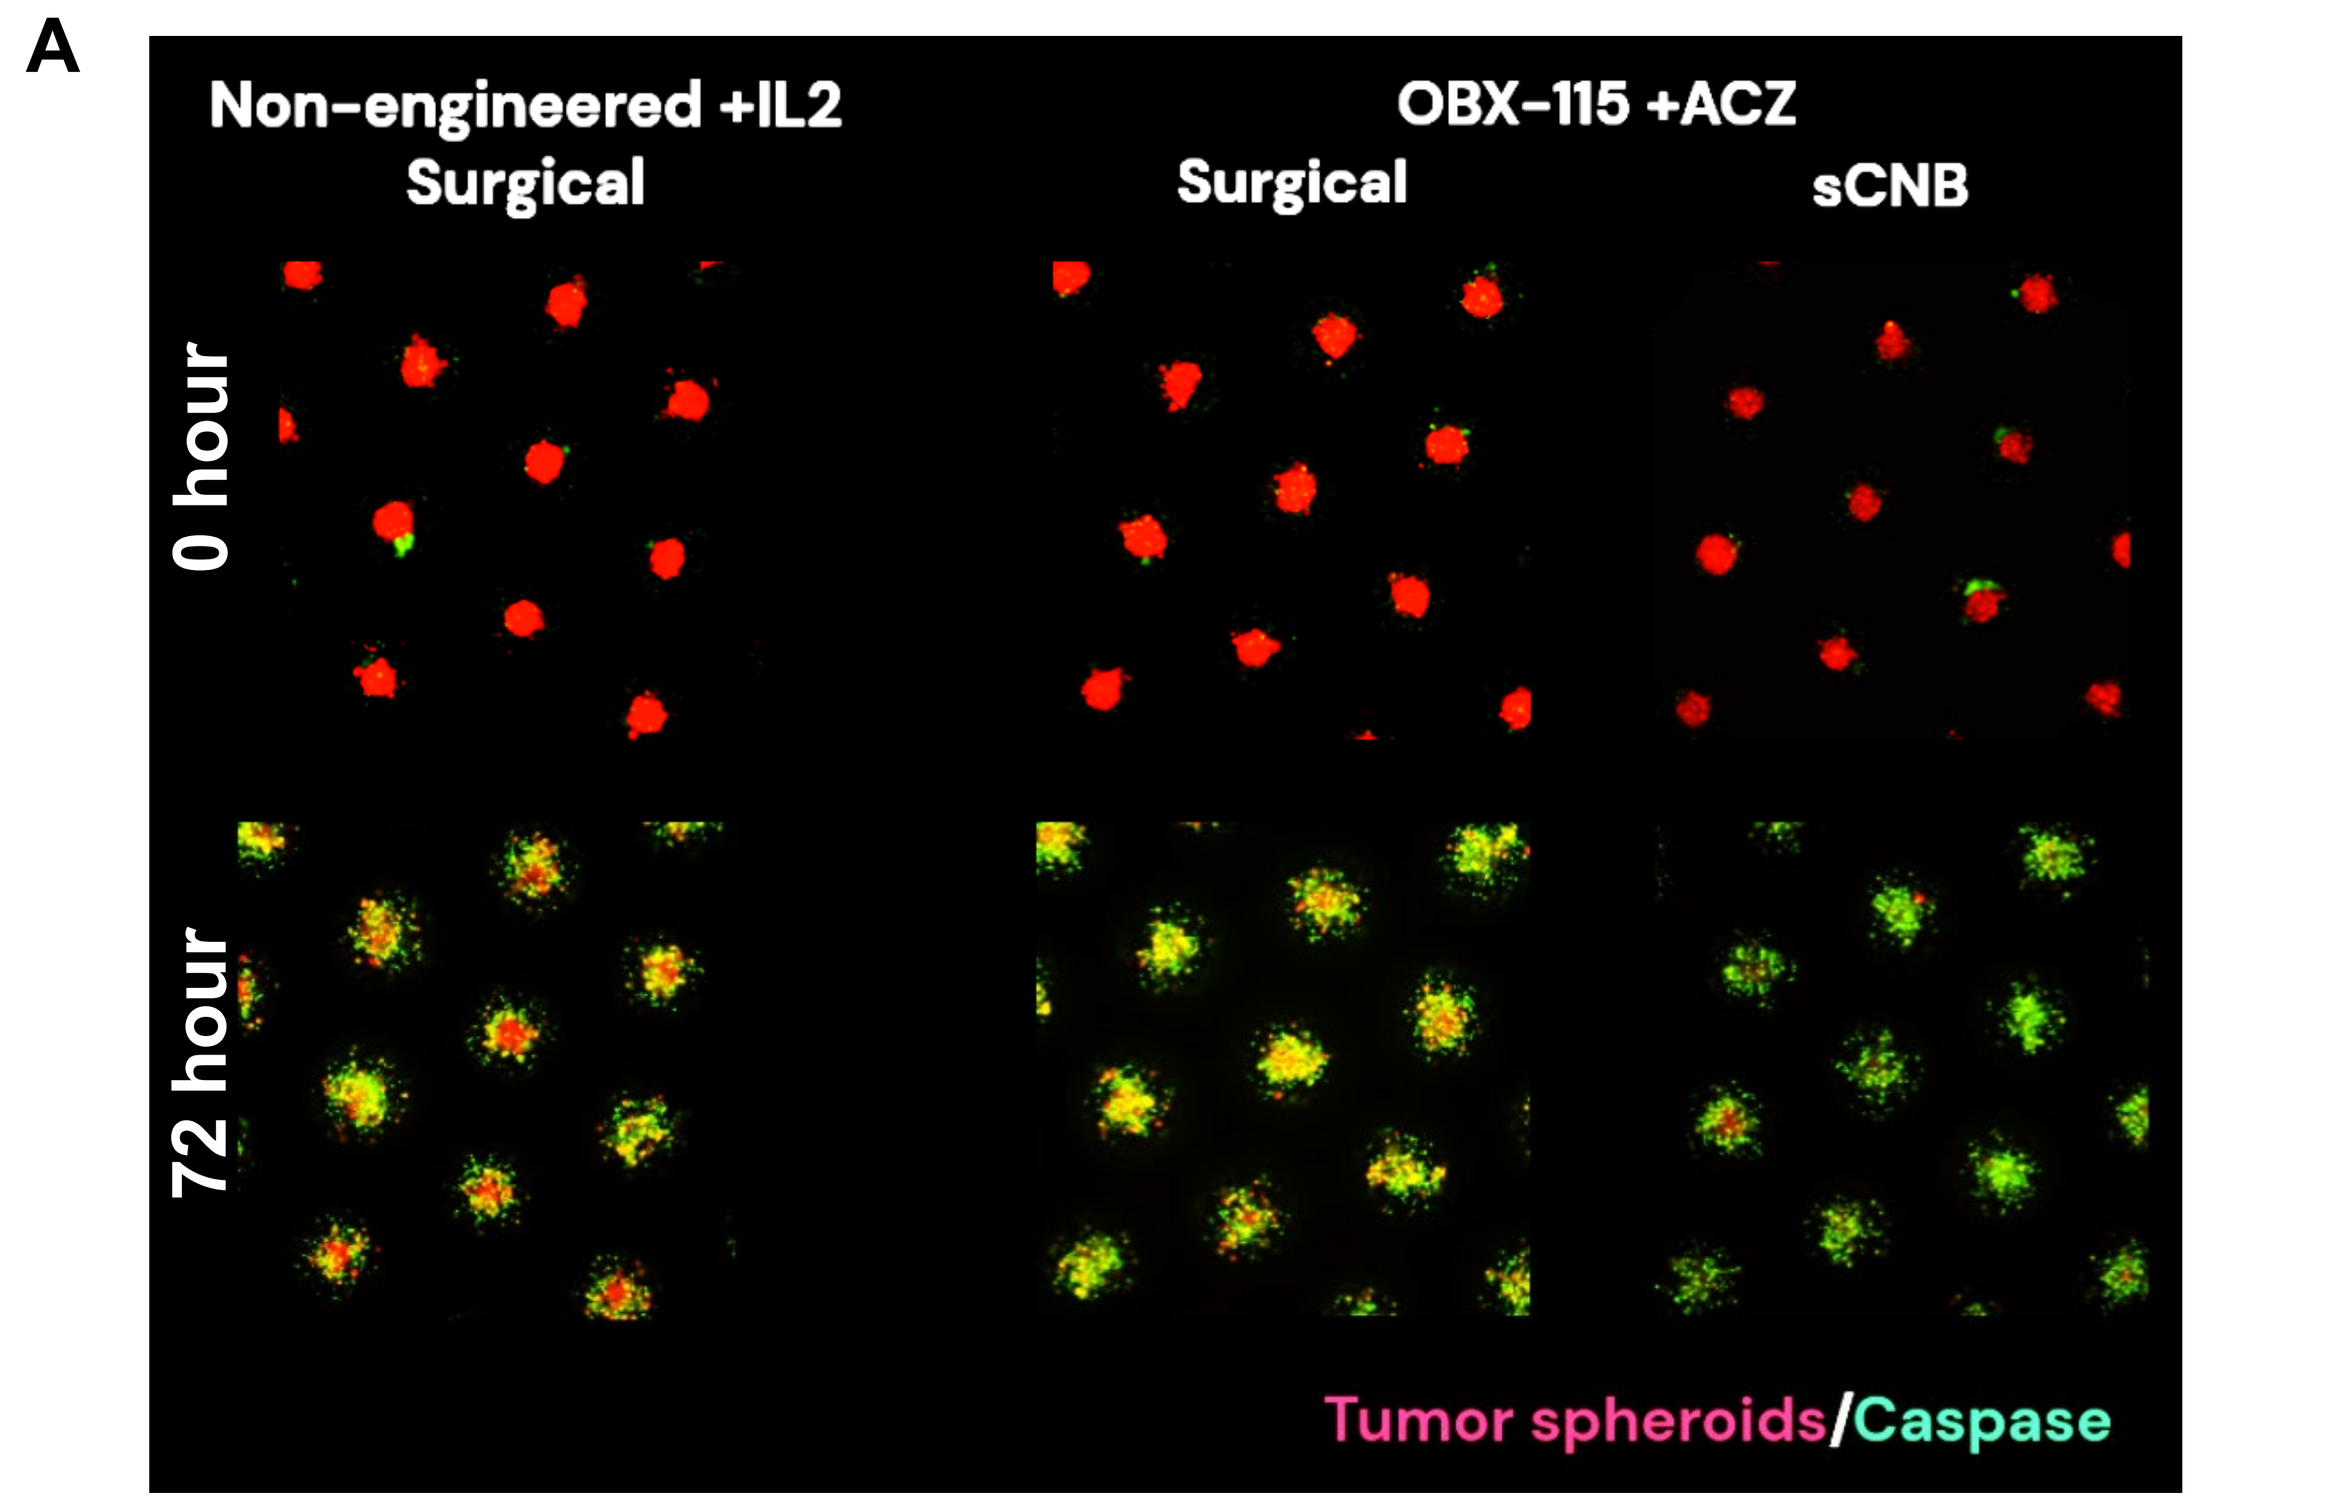
A
Non-engineered +IL2
Surgical
OBX-115 +ACZ
Surgical
sCNB
0 hour
72 hour
Tumor spheroids/Caspase

Figure 8. OBX-115 TIL generated from both surgical and sCNB tissue exert superior potent cytotoxicity against autologous tumor compared with non-engineered TIL (*continued*)

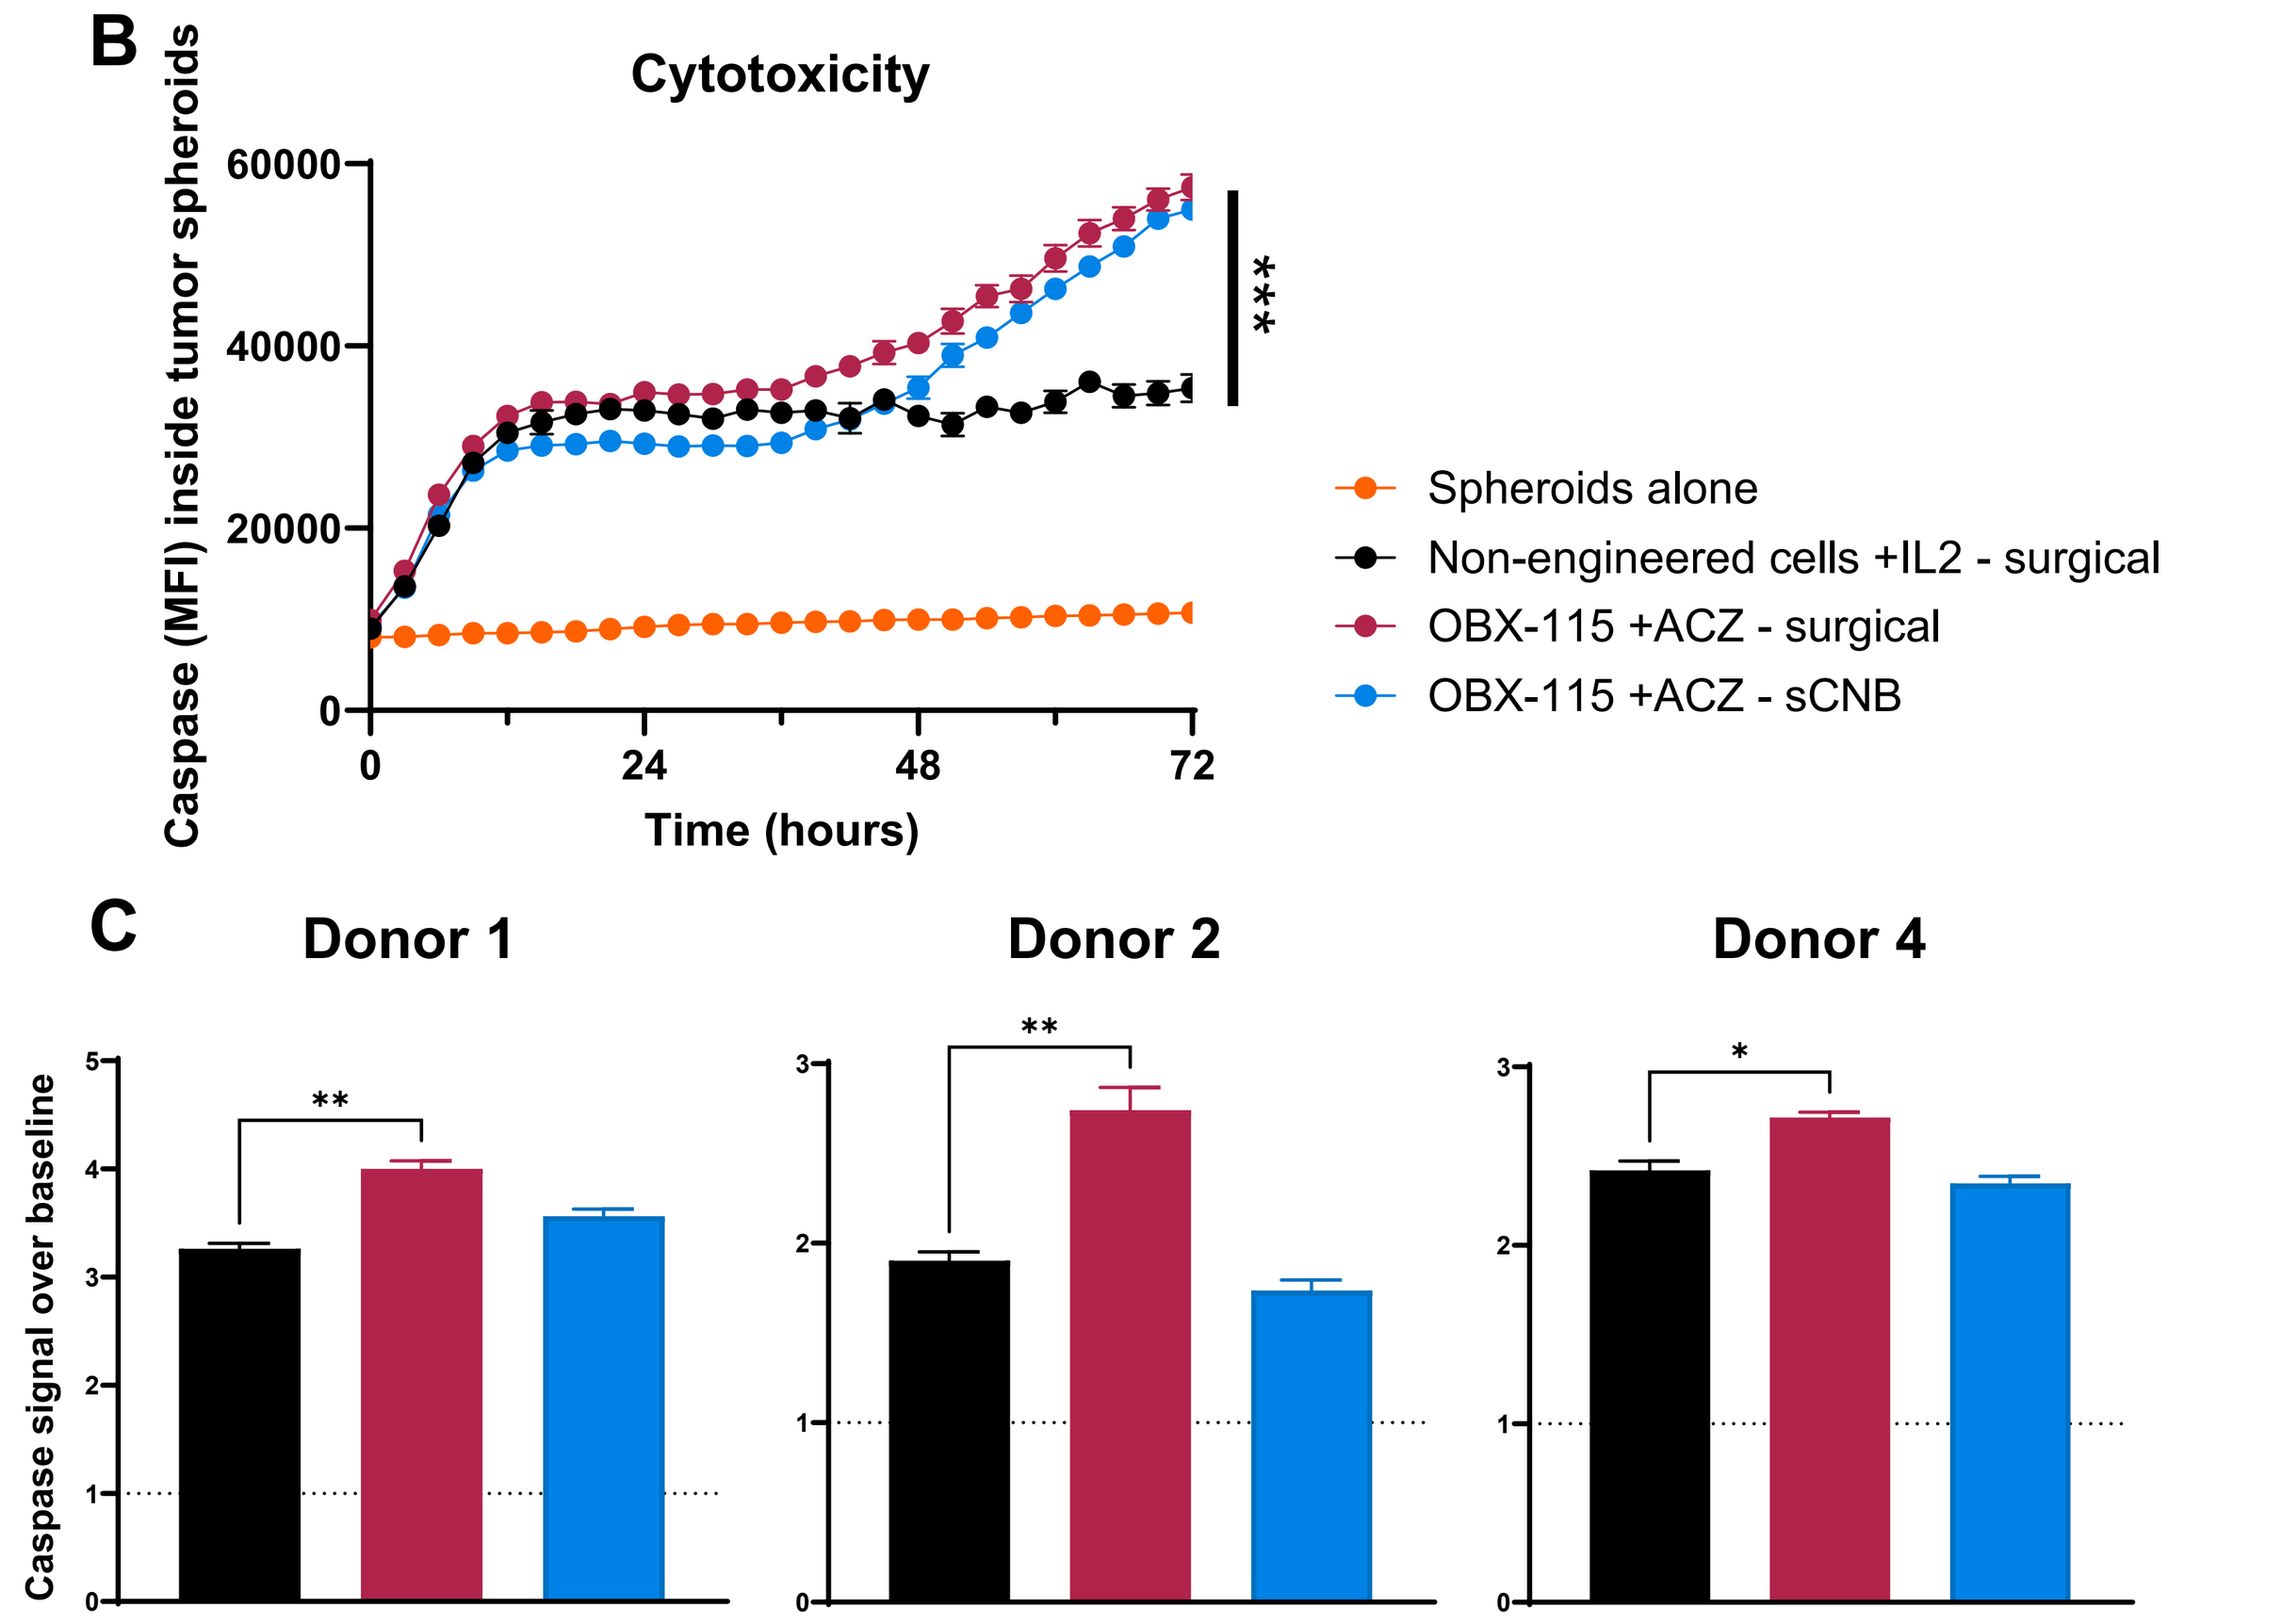
B
Cytotoxicity
Caspase (MFI) inside tumor spheroids
Time (hours)

Spheroids alone
Non-engineered cells +IL2 - surgical
OBX-115 +ACZ - surgical
OBX-115 +ACZ - sCNB
C
Donor 1
Donor 2
Donor 4
Caspase signal over baseline

Figure 8. OBX-115 TIL generated from both surgical excision and sCNB show superior cytotoxicity against autologous tumor spheroids when compared with non-engineered TIL. When co-cultured with autologous tumor spheroids (red cell tracker dye labeled), OBX-115 generated from surgical excision and sCNB tumor tissue could both effectively elicit cytotoxicity (indicated by green caspase 3/7 dye, **A–B**). Similar results were observed across all 3 donors tested, as shown by area under the curve for caspase 3/7 signal inside tumor spheroids normalized against baseline signal in spheroids alone (**C**) (*p<0.05, **p<0.01, ***p<0.005; one-way ANOVA).

Figure 9. Successful expansion of OBX-115 TIL from cCNB (Donor 5) demonstrated a similar phenotype, function, and ACZ-dependent mbIL15 signaling compared to historic surgical

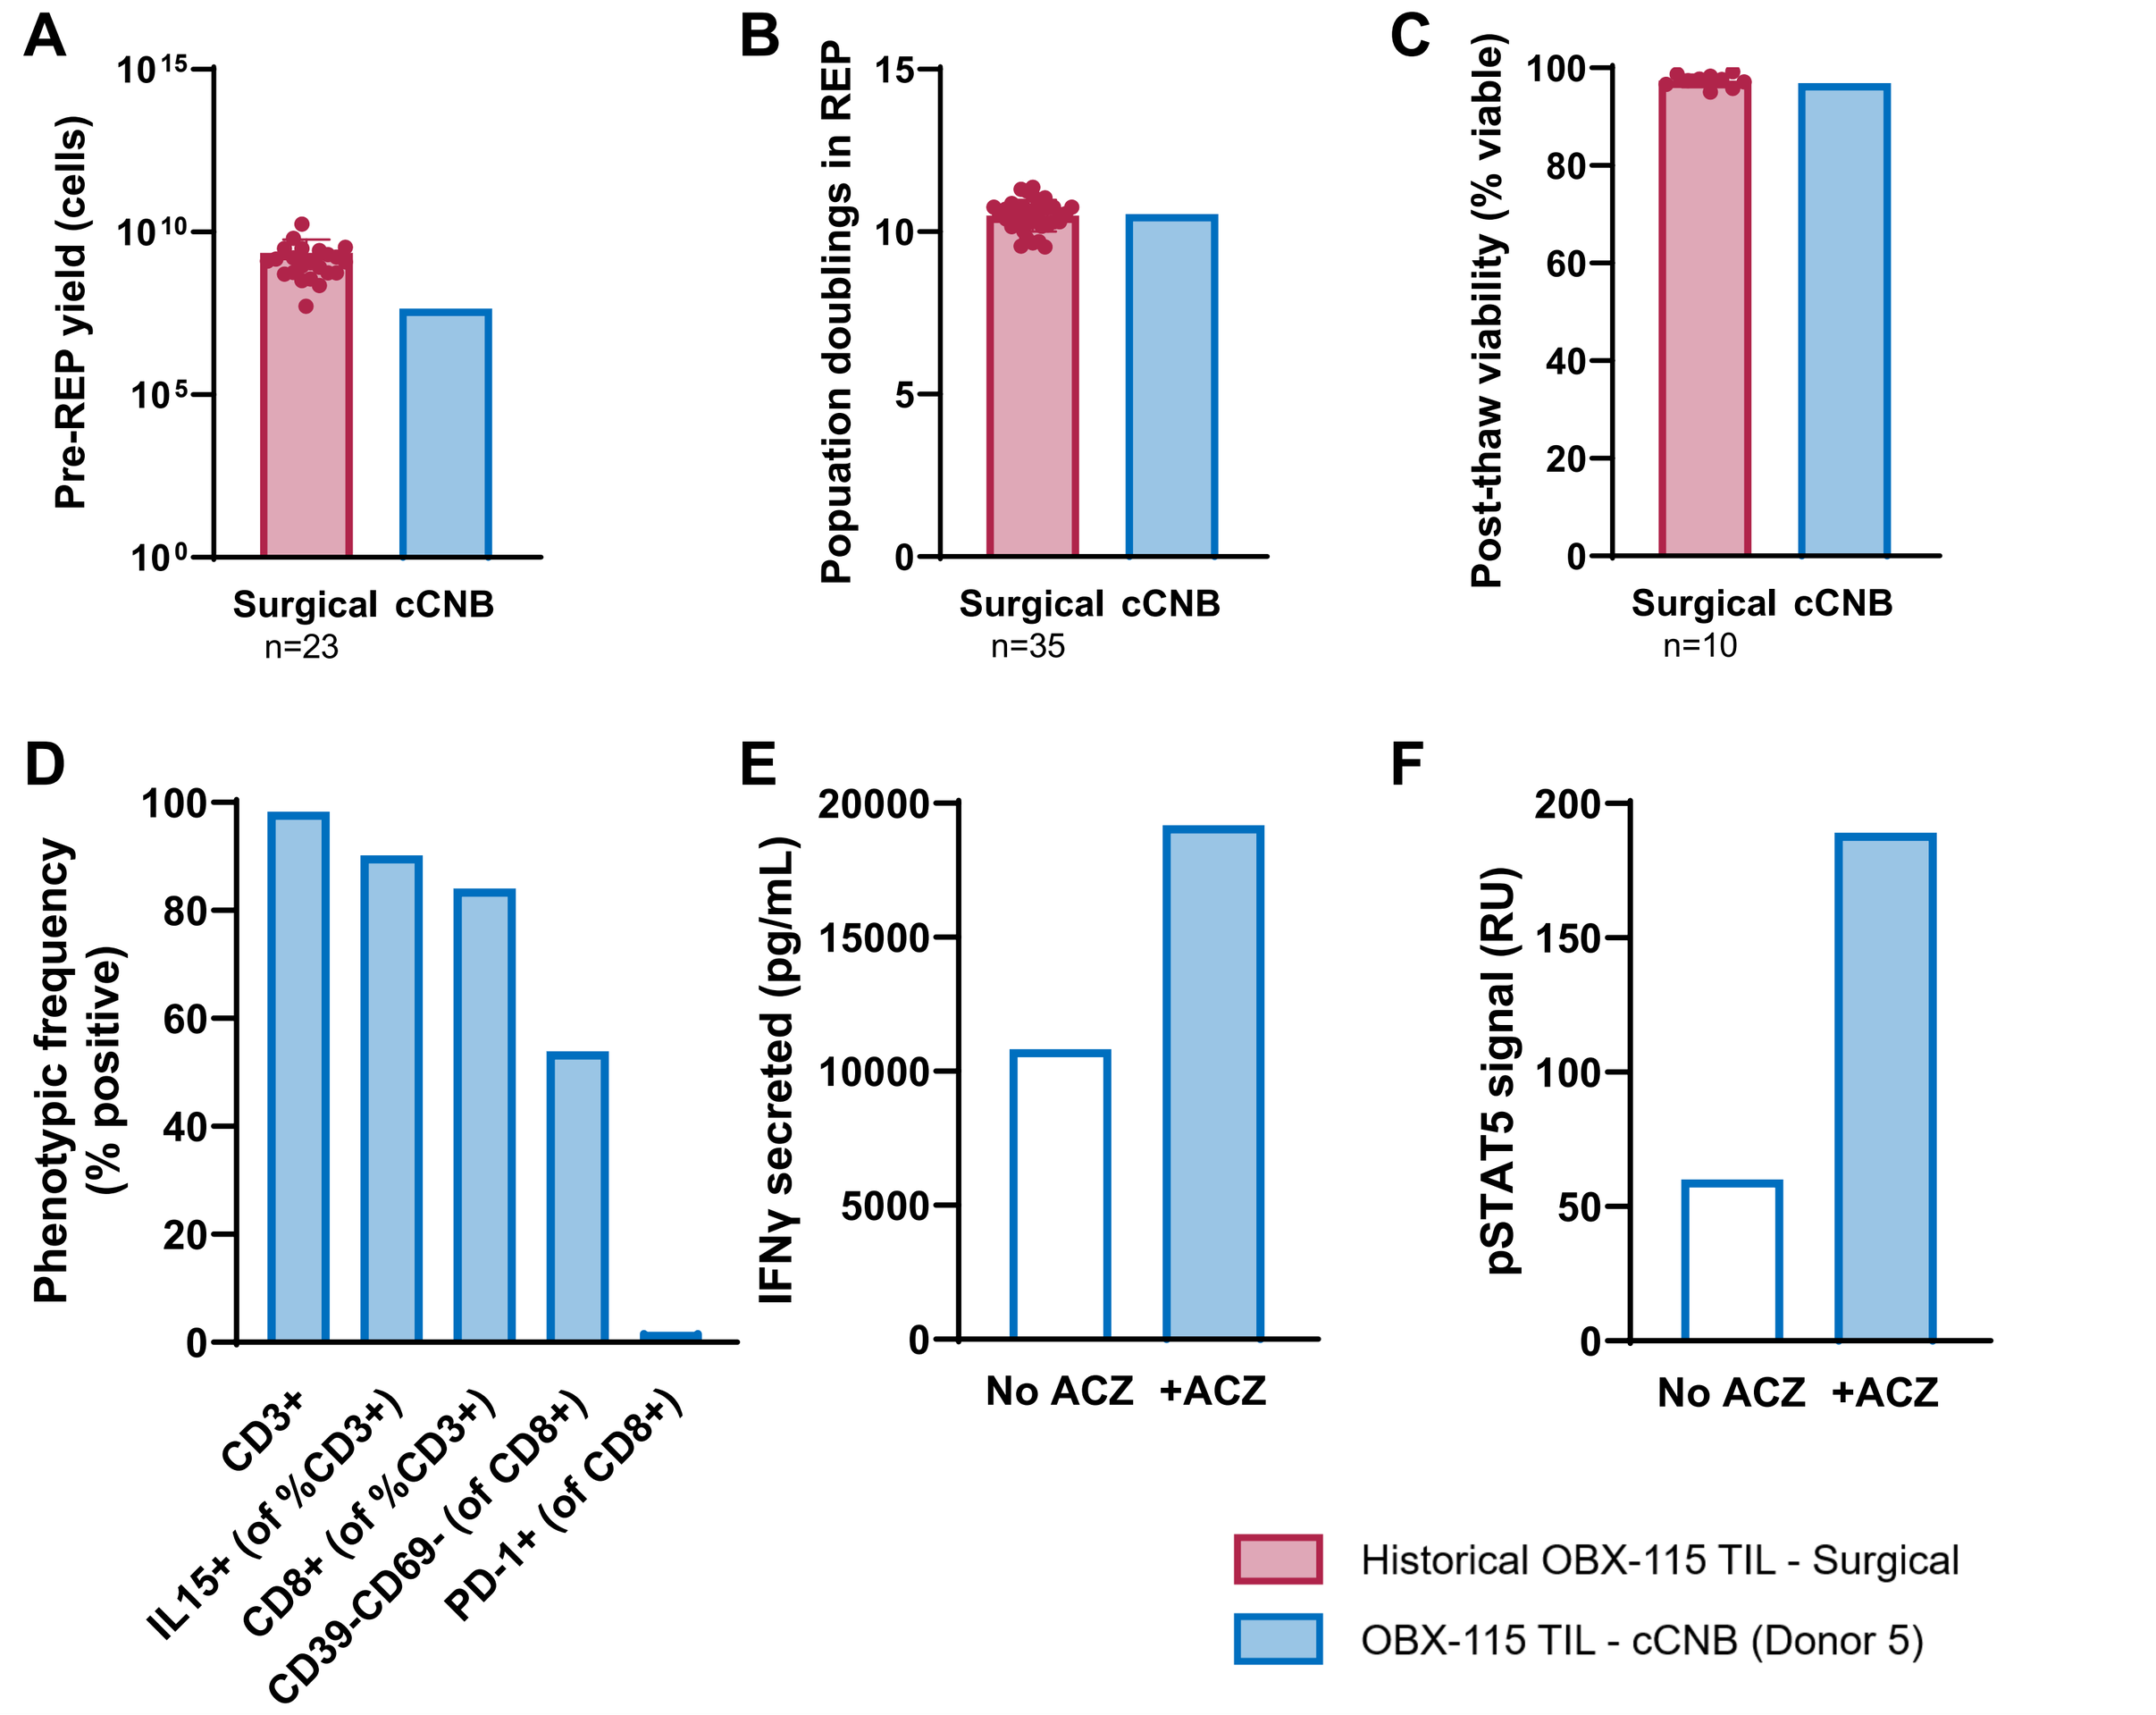
A
Pre-REP yield (cells)
Surgical cCNB
n=23
B
Popuation doublings in REP
n=35
C
Post-thaw viability (% viable)
n=10
D
Phenotypic frequency (% positive)
CD3+
IL15+ (of %CD3+)
CD8+ (of %CD3+)
CD39-CD69- (of CD8+)
PD-1+ (of CD8+)
E
IFNγ secreted (pg/mL)
No ACZ +ACZ
F
pSTAT5 signal (RU)
Historical OBX-115 TIL - Surgical
OBX-115 TIL - cCNB (Donor 5)

Figure 9. Successful expansion of OBX-115 TIL from cCNB (Donor 5) demonstrated a similar phenotype, function, and ACZ-dependent IL15 signaling compared to historic surgical (*continued*)

Figure 9. Successful expansion of OBX-115 TIL from cCNB (Donor 5) with similar product attributes to those obtained from historic surgical samples tested at PD lab. Pre-REP TIL successfully generated from IR-guided cCNB using proprietary pre-REP process (**A**). OBX-115 TIL derived from cCNB exhibited comparable growth kinetics and viability, indicating successful expansion (**B–C**). Flow cytometry analysis revealed OBX-115 TIL from cCNB tumor tissue were predominately CD8+ T cells, with high expression of DRD-regulatable mbIL15, high proportion of "stem-like" (CD8+CD39-CD69-) T cells, and low proportion of exhausted (PD-1+) T cells (**D**). Functionality of OBX-115 TIL from cCNB tumor tissue was evidenced by increased IFNγ secretion upon anti-CD3/anti-CD28 stimulation (**E**), and by increased STAT5 phosphorylation, indicative of ACZ-dependent mbIL15 signaling (**F**).

Conclusions

Previously reported clinical outcomes suggest OBX-115 manufactured using tumor tissue from core needle biopsy produced clinical responses in advanced melanoma. Here, we demonstrated that the OBX-115 manufacturing process successfully generates a **phenotypically and functionally similar OBX-115 product** across a large range of tumor tissue starting material and allows clinical core needle biopsy procurement in NSCLC:

- Pre-REP and REP **TIL expansion was similar** using surgical and core needle biopsy tumor tissue
- OBX-115 generated from paired surgical and core needle biopsy tissues have a **similar phenotype**: predominantly **effector memory**, **low proportion of PD-1** expression, and **high proportion of "stem-like" progenitor CD8+CD39-CD69-** T cells, with **similar TCR clonotype**
- OBX-115 generated from surgical excision and core needle biopsy tumor tissue show **similar polyfunctionality** upon activation and **superior cytotoxicity** against autologous tumor spheroids as compared with non-engineered TIL
- Together, these data **support tumor tissue procurement using core needle biopsy in patients with NSCLC in the ongoing Agni-01 clinical trial** (NCT06060613) in addition to the established core needle biopsy methodology for melanoma

References

1. Schoenfeld AJ, et al. Cancer Discov 2024;14(8):1389-1402.
2. Pedro, KD, et al. Cancer Research 83.8_Supplement (2023): Abstract LB096.
3. Amaria RN, et al. J Clin Oncol 2024;42(Suppl 16):Abstract 9515.
4. Bernatchez C. Cytotherapy, Volume 26, Issue 6, Supplement (June 2024): S154-S155.
5. Krishna S, et al. Science 2020;370(6522):1328-1334.

Abbreviations

ACZ, acetazolamide; cis, cisplatin; CNB, core needle biopsy; DP, drug product; IL15, interleukin 15; IR, interventional radiology; mbIL15, membrane-bound IL15; NSCLC, non-small cell lung cancer; pre-REP, pre-rapid expansion protocol; REP, rapid expansion protocol; sCNB, stimulated core biopsy; TIL, tumor-infiltrating lymphocytes; PBMC, peripheral blood mononuclear cells.

Acknowledgments

- This study is funded by Obsidian Therapeutics, Inc. (Cambridge, MA, USA)
- Editorial assistance was provided by Amanda Kelly (Obsidian Therapeutics)
- Schematics were generated with BioRender

Disclosures

MB reports a consulting relationship with Obsidian Therapeutics.

© 2024 Obsidian Therapeutics, Inc.

Presented at Society for Immunotherapy of Cancer 39th Annual Meeting | November 6–10 | Houston, TX

obsidiantx.com